etrolizumab

P1607

Etrolizumab Treatment Increases Blood Levels of CD3, Treg and Th17 Cells Measured Through Epigenetic Activation of CD3, FOXP3 and IL-17 Loci

Franklin Fuh,[1] Eva Raschke,[2] Akiko Chai,[1] Caroline Looney,[1] Thomas O. Kleen,[2] Séverine Vermeire,[3] Sharon O'Byrne,[1] Teresa Ramirez-Montagut[1]

[1]Genentech Research and Early Development, South San Francisco, CA, United States; [2]Epiontis GmbH, Berlin, Germany; [3]Division of Gastroenterology, University of Leuven, Leuven, Belgium

INTRODUCTION

- In ulcerative colitis (UC), excessive T-cell trafficking to the gut mucosa significantly contributes to intestinal inflammation[1]
- Etrolizumab is a humanized IgG1 monoclonal antibody that selectively targets the β7 subunit of the α4β7 and αEβ7 integrins with high affinity and blocks interactions with their respective ligands, mucosal addressin cell adhesion molecule-1 (MAdCAM-1) and E-cadherin **(Figure 1)**

Figure 1. Mechanism of Action of Etrolizumab

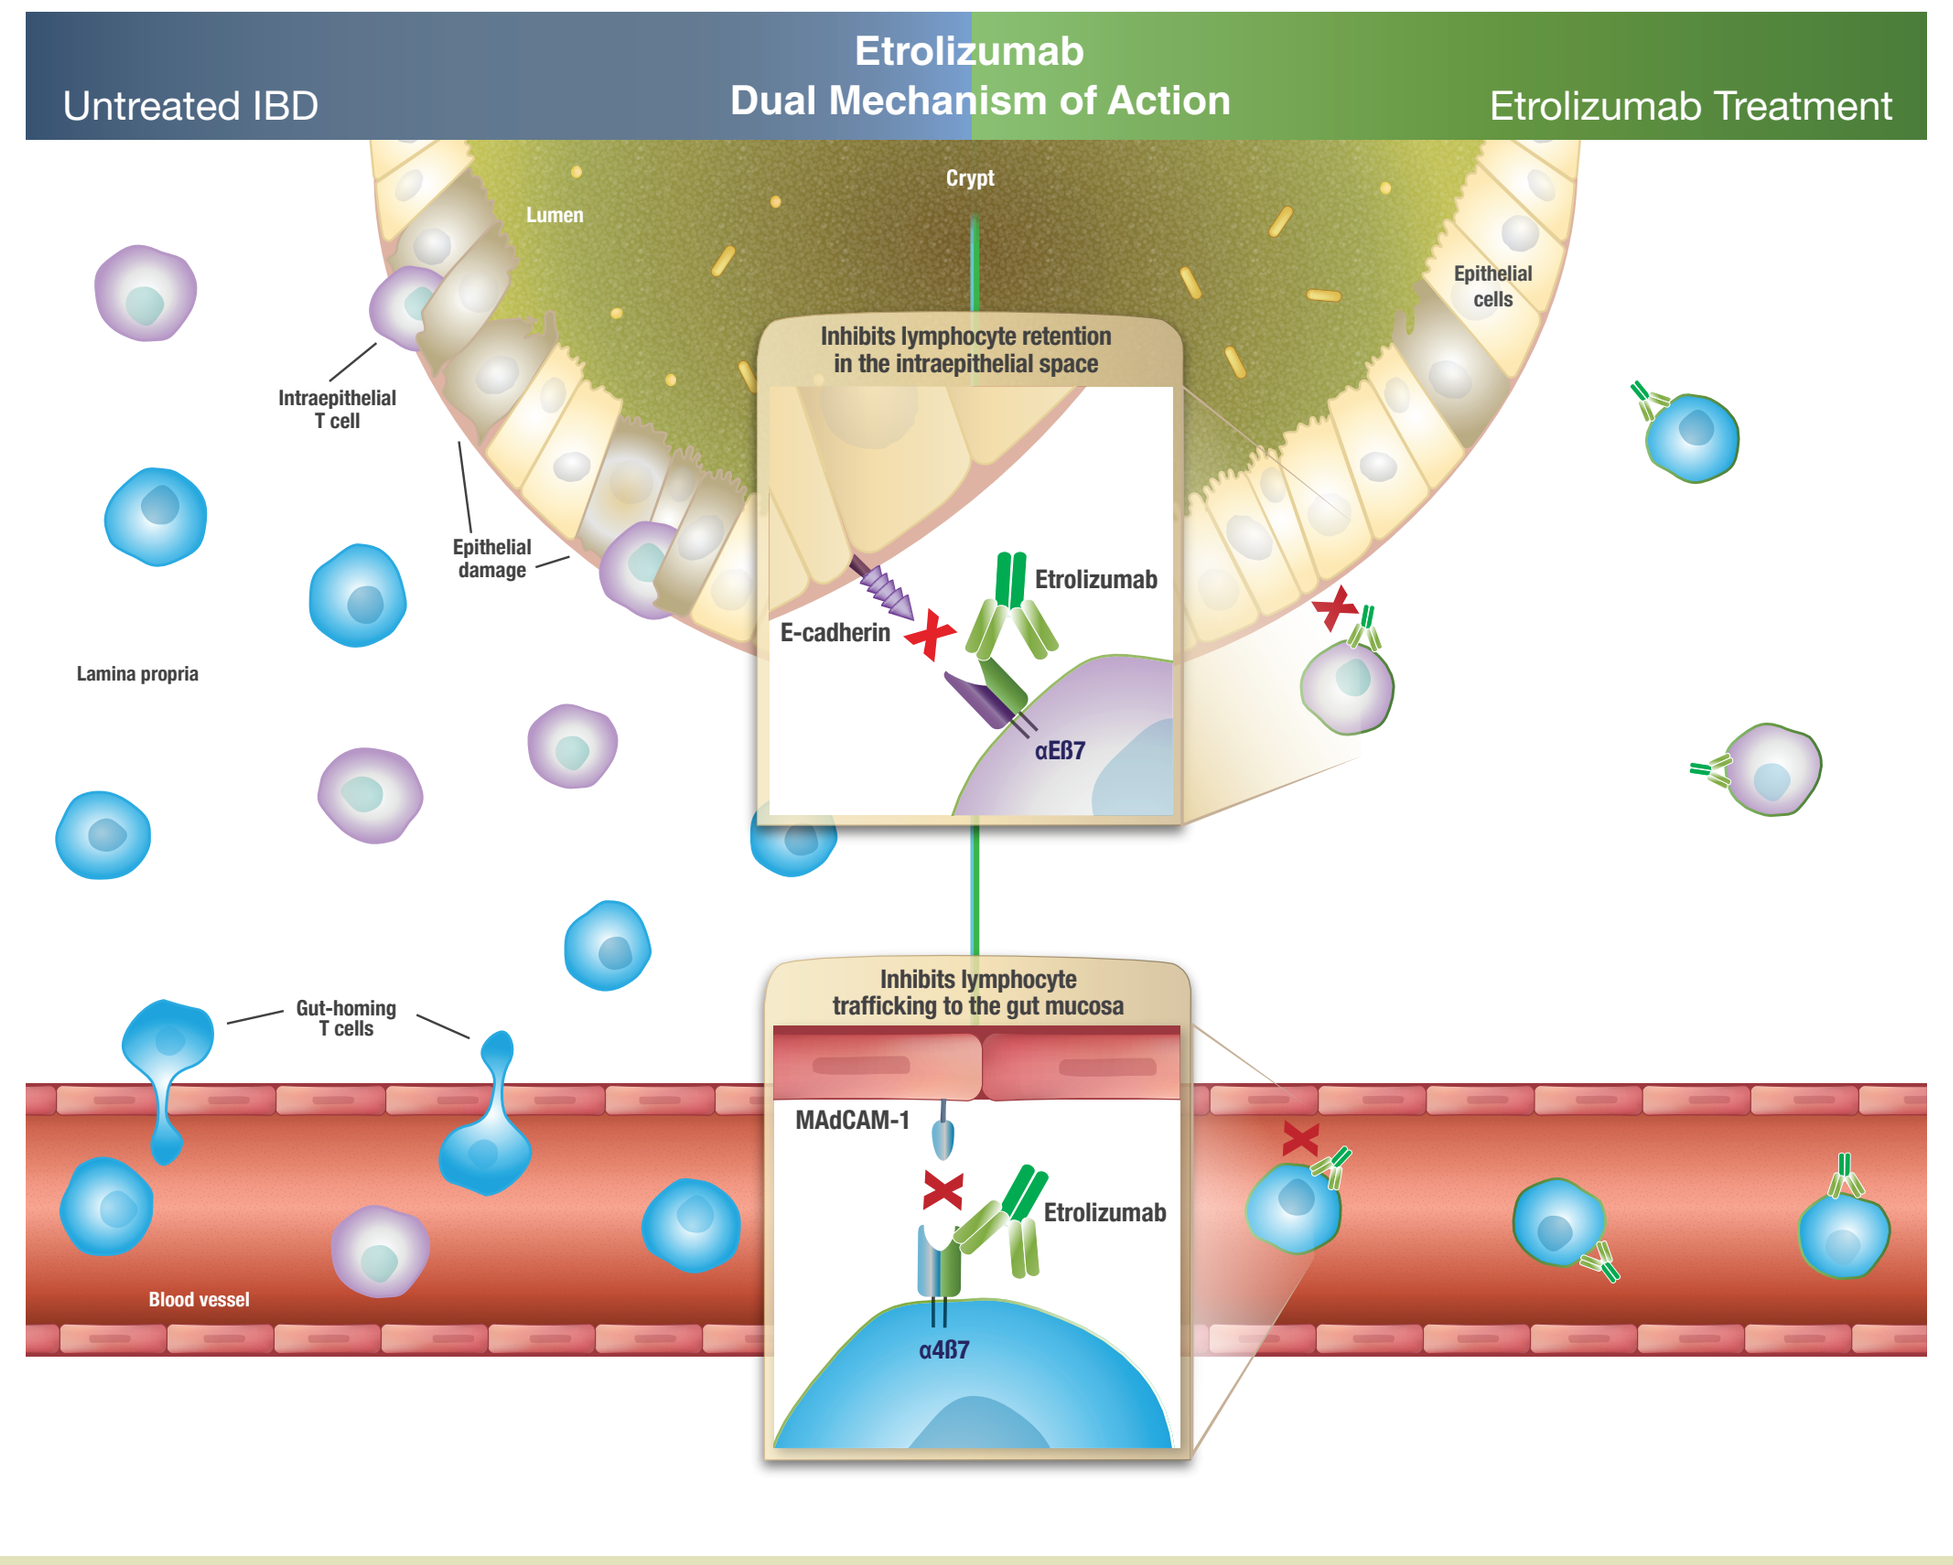

Untreated IBD
Etrolizumab
Dual Mechanism of Action
Etrolizumab Treatment
Crypt
Lumen
Epithelial cells
Intraepithelial T cell
Epithelial damage
Lamina propria
Inhibits lymphocyte retention in the intraepithelial space
E-cadherin
Etrolizumab
αEβ7
Gut-homing T cells
Inhibits lymphocyte trafficking to the gut mucosa
MAdCAM-1
Etrolizumab
α4β7
Blood vessel

- The Phase 2 EUCALYPTUS study demonstrated that patients treated with etrolizumab for 10 weeks achieved a significantly higher rate of remission induction compared with patients receiving placebo
- Analyses of patient subsets and lymphocyte dynamics in EUCALYPTUS highlighted the potential impact of etrolizumab on pathophysiology and disease mechanisms
- Etrolizumab-treated patients with higher than median expression levels of αE integrin in baseline colonic biopsies demonstrated higher remission rates, suggesting that αE may be a biomarker that predicts greatest benefit from etrolizumab treatment[2]
- Significant increases in the number of intestinal homing β7+ T lymphocytes in the peripheral blood were observed in patients treated with etrolizumab compared with placebo-treated patients, demonstrating the impact of etrolizumab treatment in reducing T-cell traffic to the intestinal mucosa[2]

OBJECTIVES

- In this analysis of EUCALYPTUS, we further assessed the effect of etrolizumab on levels of peripheral blood T lymphocytes and T-lymphocyte subsets using a novel approach
 - To quantify the number of CD3+ T cells
 - To examine forkhead box P3 (FOXP3)+ regulatory T cells (Treg; these cells help to maintain gut homeostasis) and T helper 17 cells (Th17; these cells are implicated in the pathogenesis of inflammatory bowel disease [IBD])[3]

METHODS

- Blood samples were obtained from healthy controls (n = 60) and from patients with UC enrolled in the EUCALYPTUS study at day 1 pre-dose and at days 15, 29, 43 and 71 post-dose
 - EUCALYPTUS is a global, randomized, double-blind, placebo-controlled multicenter Phase 2 study in which patients with moderately to severely active UC who had failed conventional therapy (n = 124) were randomized in a 1:1:1 ratio to subcutaneous injections of etrolizumab 100 mg (on weeks 0, 4 and 8, with placebo at week 2), etrolizumab 300 mg (on weeks 2, 4 and 8 plus a 420-mg loading dose at week 0) or matching placebo **(Figure 2)**

Figure 2. EUCALYPTUS Study Design

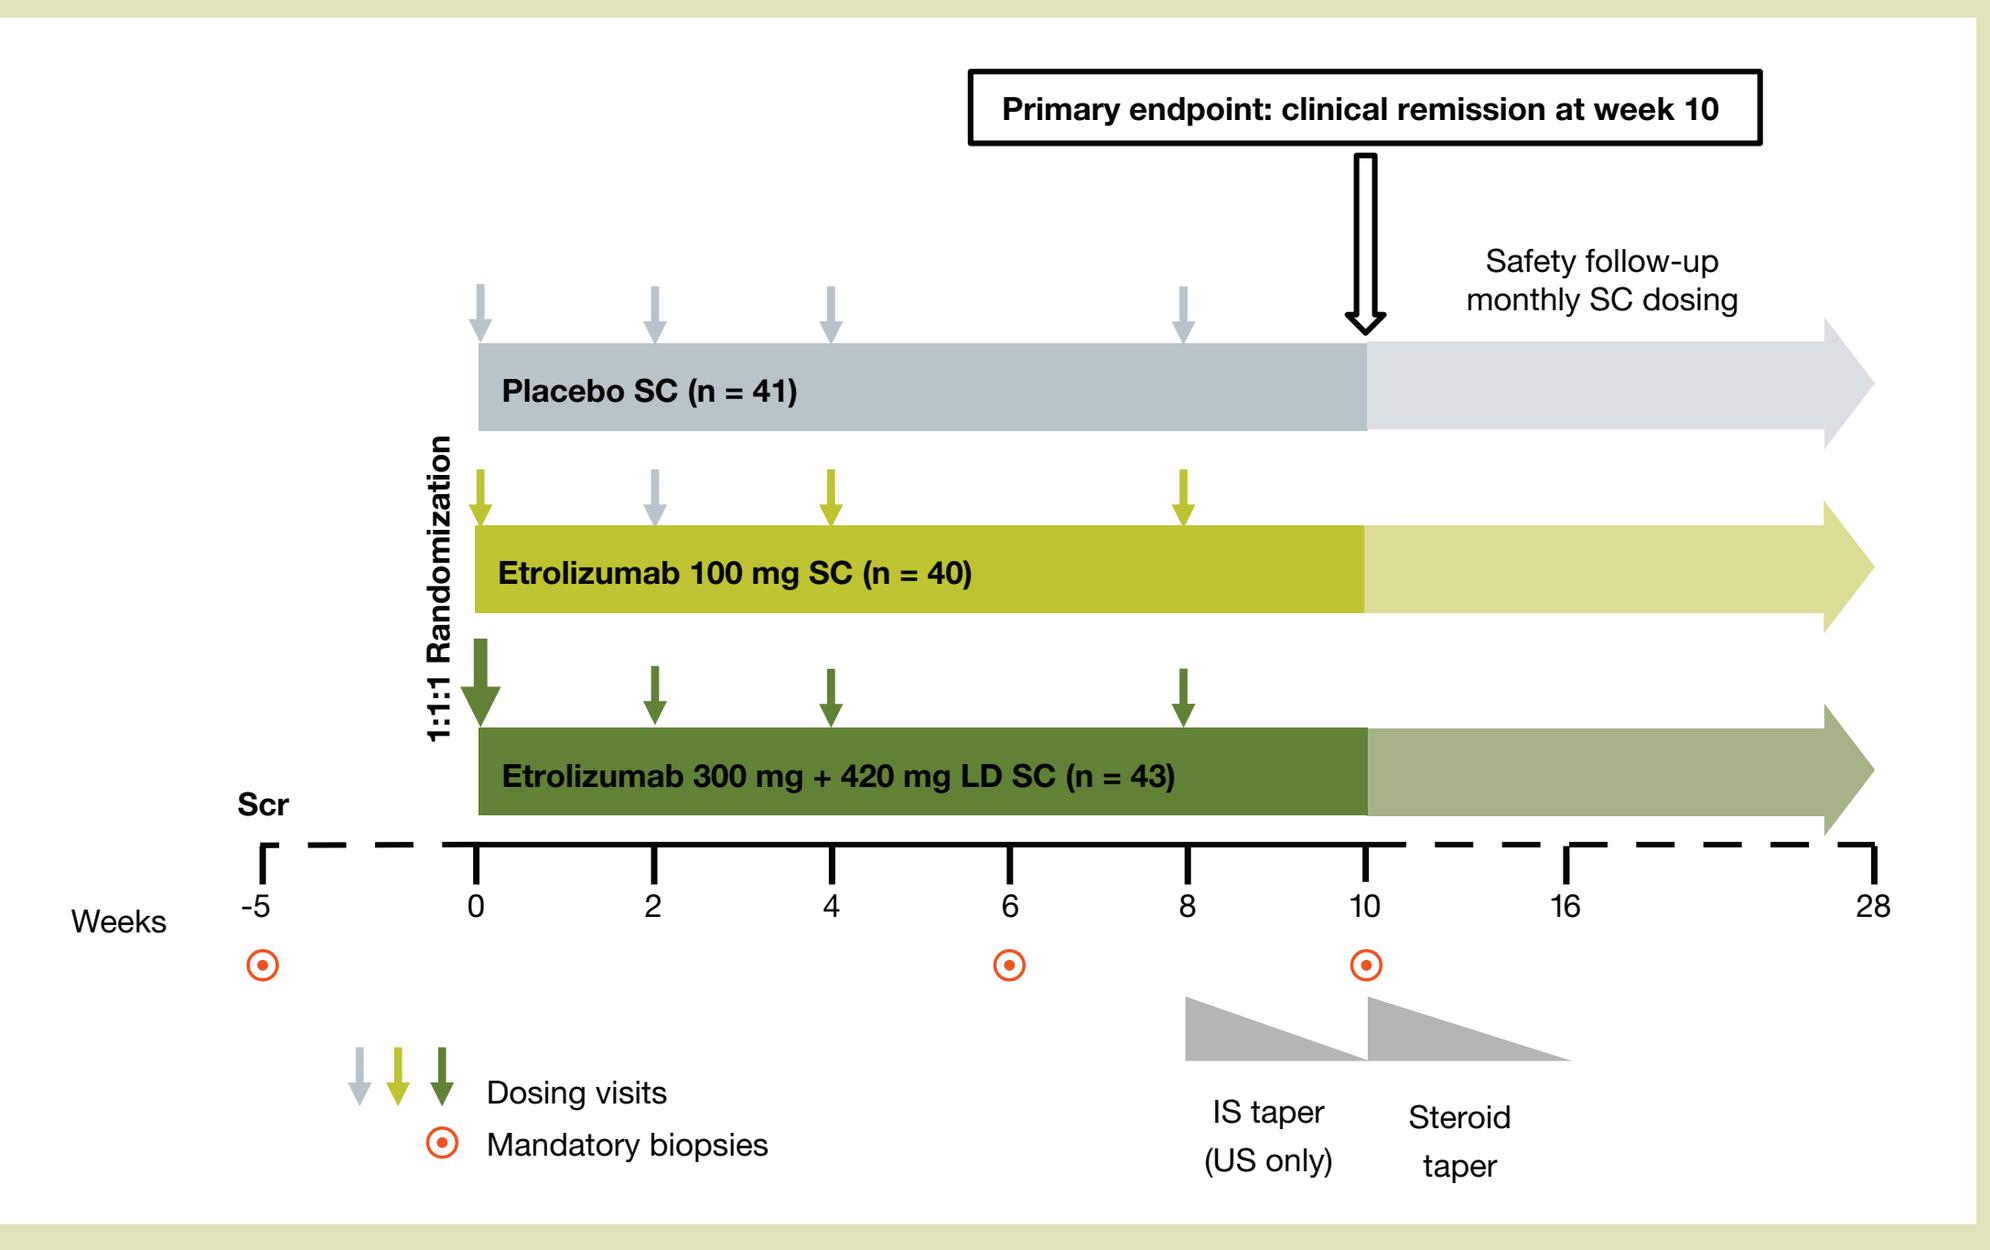

Primary endpoint: clinical remission at week 10
Safety follow-up monthly SC dosing
Placebo SC (n = 41)
Etrolizumab 100 mg SC (n = 40)
Etrolizumab 300 mg + 420 mg LD SC (n = 43)
1:1:1 Randomization
Scr
Weeks
-5
0
2
4
6
8
10
16
28
Dosing visits
Mandatory biopsies
IS taper (US only)
Steroid taper

IS, immunosuppressant; LD, loading dose; SC, subcutaneous; Scr, screening. Clinical remission defined as total Mayo Clinic score ≤ 2 with no individual sub-score > 1. Actual drug amounts for the nominal 100 mg and 300 mg doses were 105 mg (1 injection) and 315 mg (3 injections), respectively (150 mg/mL, 0.7 mL per injection).

- The number of peripheral blood total T cells (CD3+) and those committed to Treg or Th17 lineages were assessed by measuring epigenetic activation (demethylation) of the CD3, FOXP3 and IL-17 loci, respectively
 - Epigenetic activation at all 3 loci has been previously shown to correlate with the respective immune cell content[4-6]
 - Epigenetic activation has been shown within the *FOXP3* gene exclusively in Treg cells and not in FOXP3+ effector T cells[7]
 - Genomic DNA was isolated from whole blood samples using the DNeasy Blood & Tissue Kit (Qiagen)
 - The epigenetic quantitative polymerase chain reaction (qPCR) method is shown in **Figure 3**
 - Bisulfite conversion (BSC) was performed as previously described[4]
 - qPCR was performed as previously described for T cells,[5] Treg[4,7] and Th17 cells[6]
 - Only demethylated cytosines in the target cell type (i.e., CD4+ T cells) convert to uracil during BSC. This allows BSC genomic DNA–specific segregating primer and probes to bind and lead to a qPCR product **(Figure 3A)**
 - Methylated cytosines in nontarget cells do not change during BSC treatment of genomic DNA, leading to a primer mismatch and no qPCR product **(Figure 3B)**
 - Detection of epigenetically active (demethylated) *CD3* gene copies by qPCR determines gene copies originating from T cells **(Figure 3C)**
 - Parallel measurement of an epigenetic reference system for total cells based on epigenetically active (demethylated) GAPDH gene copies determines total cells, thus allowing robust and precise determination of relative T-cell content in the sample

Figure 3. Epigenetic Cell Counting

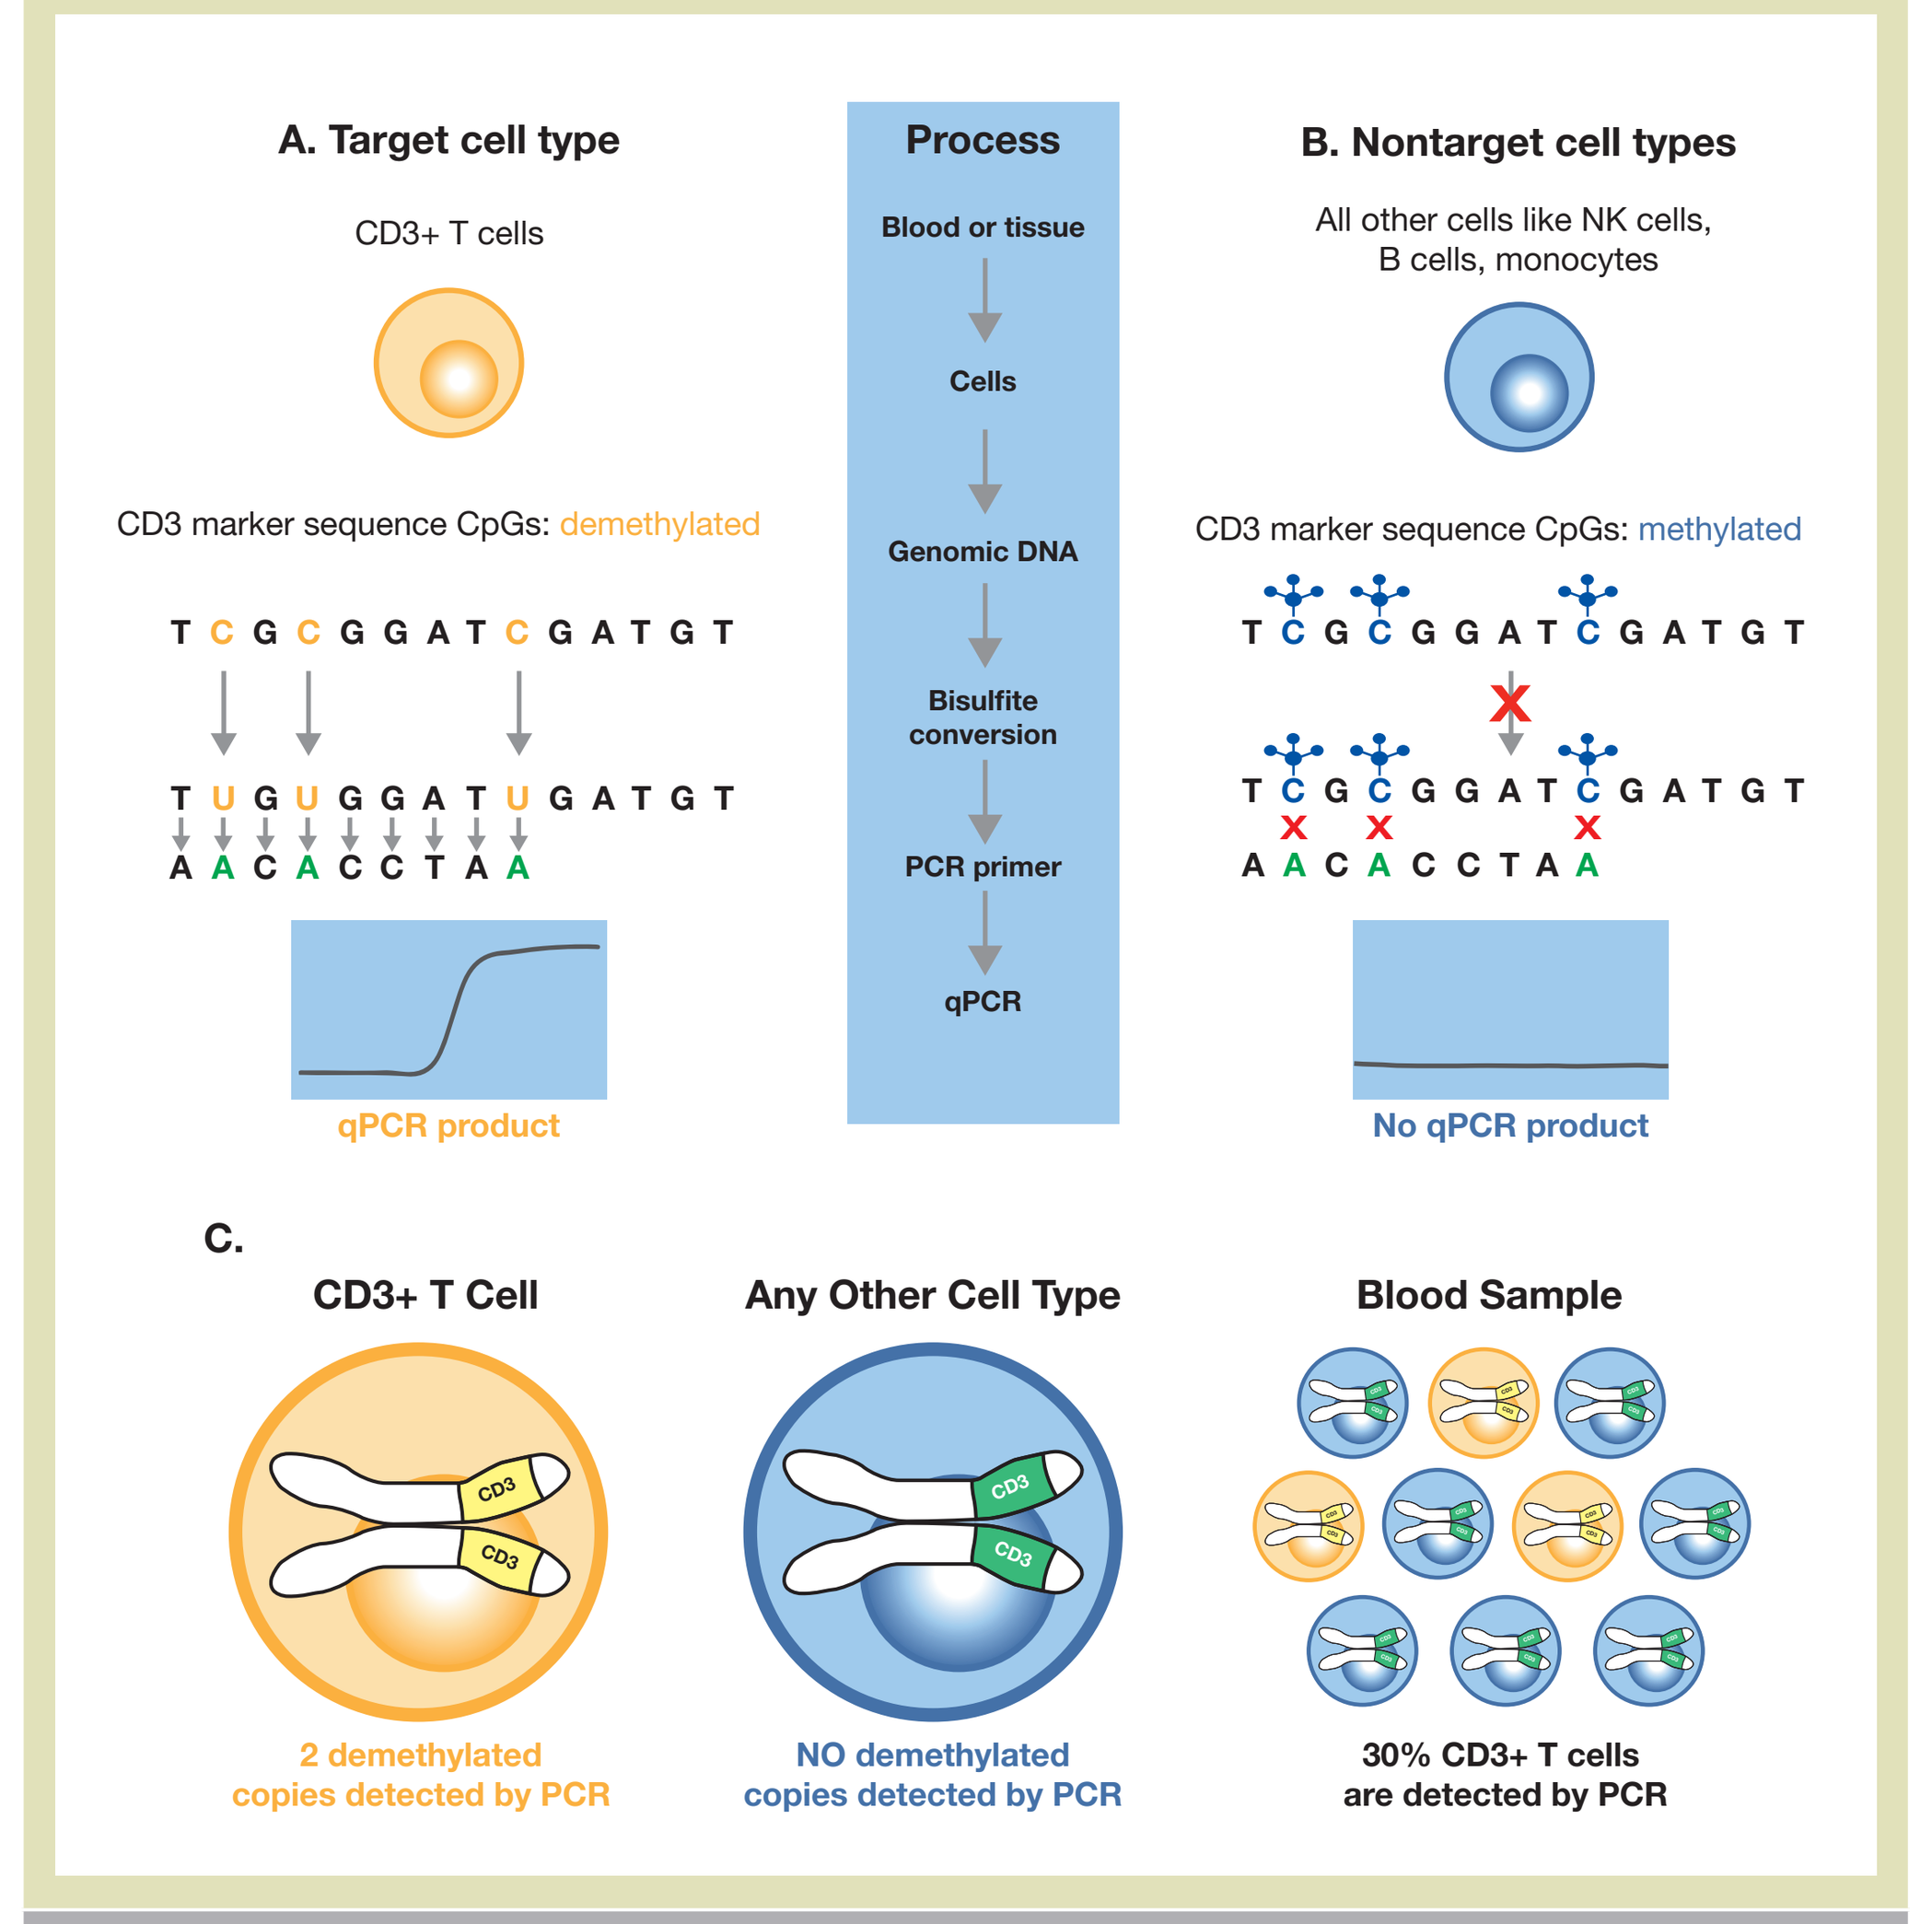

A. Target cell type
CD3+ T cells
CD3 marker sequence CpGs: demethylated
T C G C G G A T C G A T G T
T U G U G G A T U G A T G T
A A C A C C T A A
qPCR product
Process
Blood or tissue
Cells
Genomic DNA
Bisulfite conversion
PCR primer
qPCR
B. Nontarget cell types
All other cells like NK cells, B cells, monocytes
CD3 marker sequence CpGs: methylated
T C G C G G A T C G A T G T
T C G C G G A T C G A T G T
A A C A C C T A A
No qPCR product
C.
CD3+ T Cell
2 demethylated copies detected by PCR
Any Other Cell Type
NO demethylated copies detected by PCR
Blood Sample
30% CD3+ T cells are detected by PCR

NK, natural killer; qPCR, quantitative polymerase chain reaction.

RESULTS

- Patients with UC had reduced CD3+ T-cell and Treg levels compared with healthy controls **(Figure 4)**
- The comparative reduction of these cell populations in patients with UC was more pronounced in anti–tumor necrosis factor (TNF)–experienced patients

Figure 4. Lower Levels of Peripheral (A) CD3+ T cells and (B) Treg in Patients with UC Compared with Healthy Controls

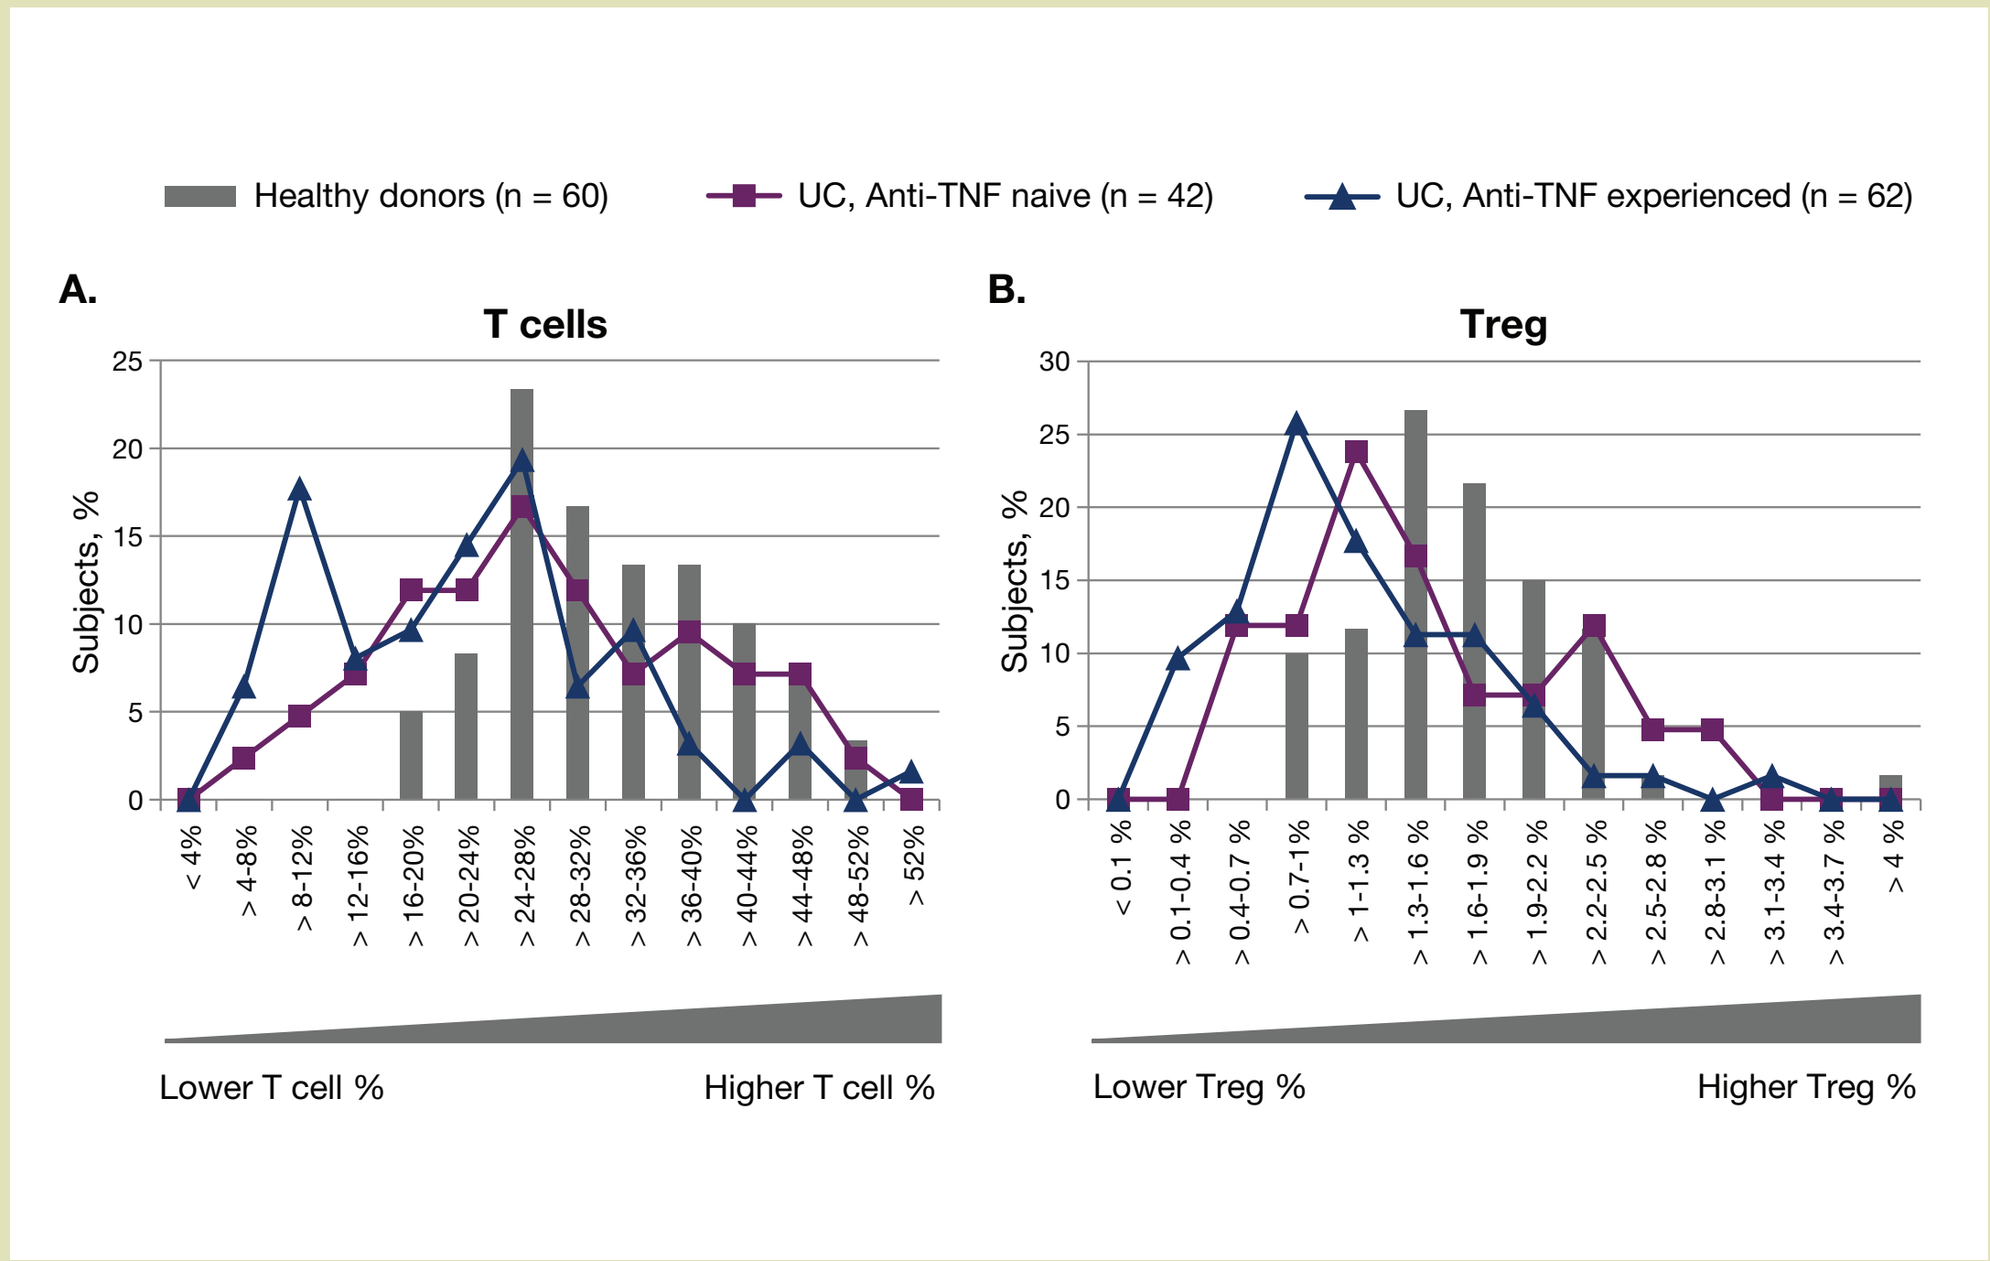

Healthy donors (n = 60)
UC, Anti-TNF naive (n = 42)
UC, Anti-TNF experienced (n = 62)
A.
T cells
Subjects, %
Lower T cell %
Higher T cell %
B.
Treg
Subjects, %
Lower Treg %
Higher Treg %

TNF, tumor necrosis factor; Treg, regulatory T cell; UC, ulcerative colitis. Data are expressed as a percentage of total number of subjects across healthy controls, anti-TNF–naive patients with UC and anti-TNF–experienced patients with UC.

- A trend toward increased levels of CD3+ T cells, Treg and Th17 cells in the peripheral blood was observed during treatment in etrolizumab-treated patients compared with placebo-treated patients **(Figure 5)**

Figure 5. Increases in (A) CD3+ T Cells, (B) Treg and (C) Th17 Cells in Etrolizumab-Treated Patients

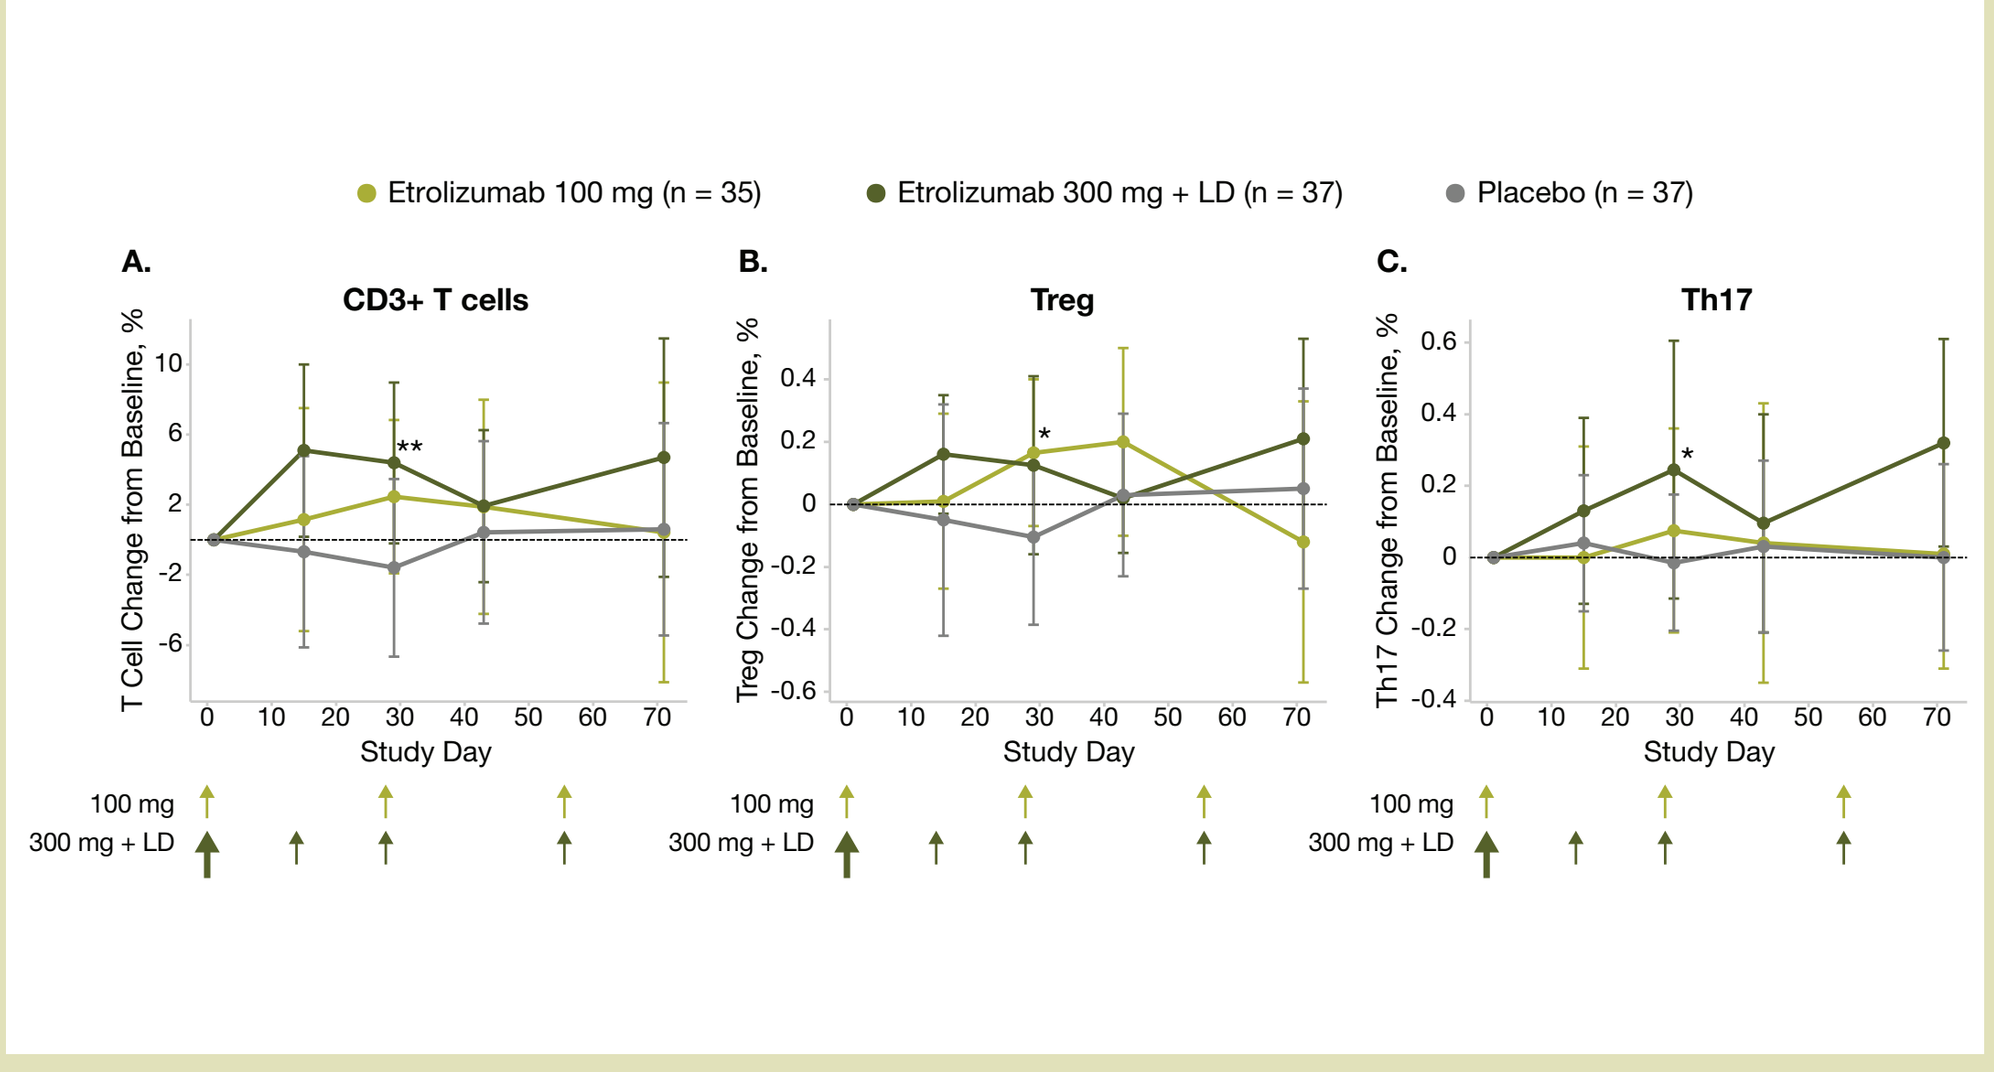

Etrolizumab 100 mg (n = 35)
Etrolizumab 300 mg + LD (n = 37)
Placebo (n = 37)
A.
CD3+ T cells
T Cell Change from Baseline, %
B.
Treg
Treg Change from Baseline, %
C.
Th17
Th17 Change from Baseline, %
Study Day
100 mg
300 mg + LD

LD, loading dose; Th17, T helper 17 cell; Treg, regulatory T cell. Data are expressed as group median ± median absolute deviation. *$P \leq 0.05$ and **$P \leq 0.02$ for etrolizumab-treated patients vs placebo-treated patients.

- Remission induced by etrolizumab treatment was associated with an overall increase in peripheral blood levels of CD3+ T cells, Treg and Th17 cells compared with baseline **(Figure 6)**
- There was no change in T-cell levels in placebo-treated patients

Figure 6. Remission in Etrolizumab-Treated Patients Was Associated with Increased Levels of (A) CD3+ T Cells, (B) Treg and (C) Th17 Cells

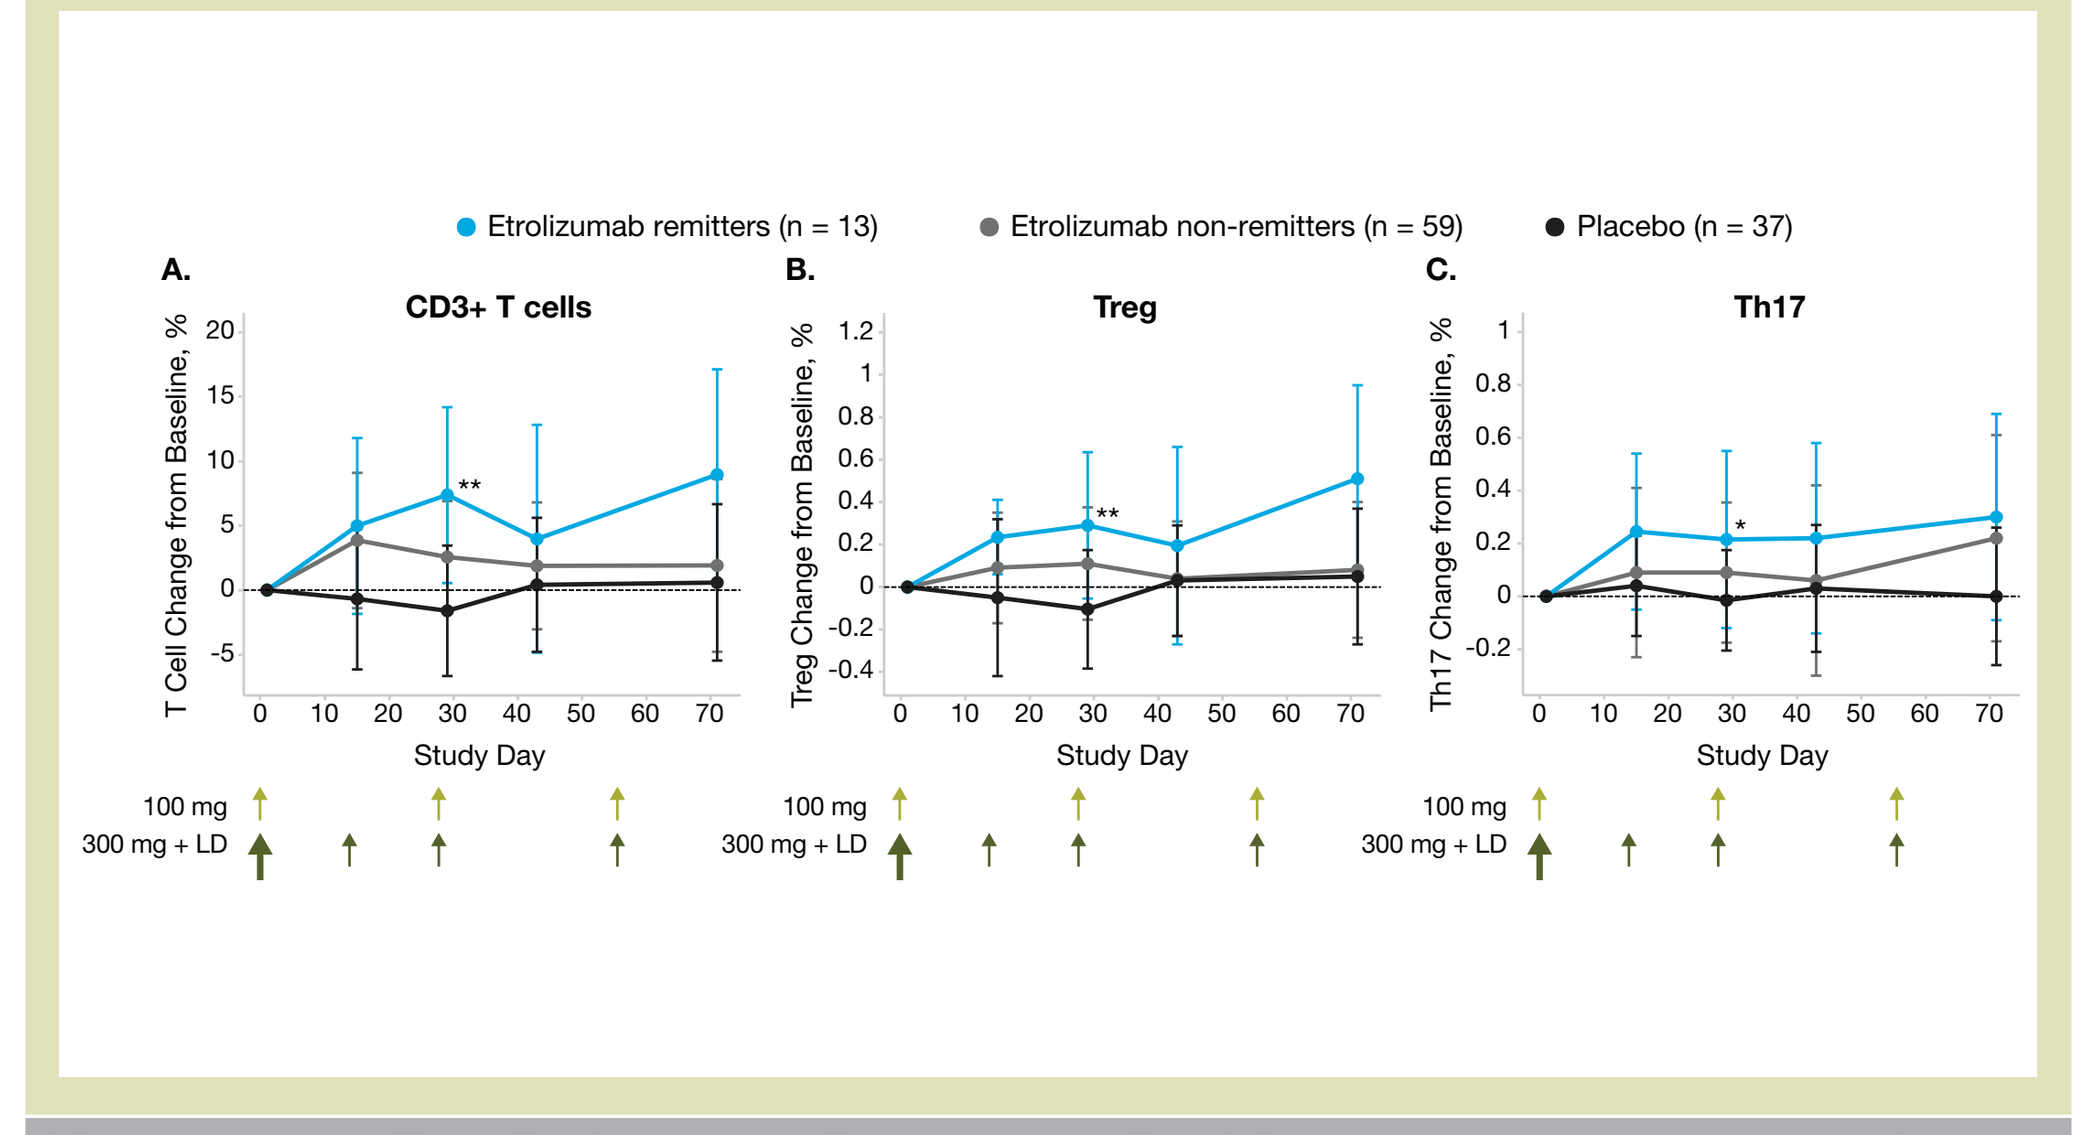

Etrolizumab remitters (n = 13)
Etrolizumab non-remitters (n = 59)
Placebo (n = 37)
A.
CD3+ T cells
T Cell Change from Baseline, %
B.
Treg
Treg Change from Baseline, %
C.
Th17
Th17 Change from Baseline, %
Study Day
100 mg
300 mg + LD

LD, loading dose; Th17, T helper 17 cell; Treg, regulatory T cell. Data are expressed as group median ± median absolute deviation. There were no remitters in the placebo arm. *$P \leq 0.05$ and **$P \leq 0.02$ for etrolizumab remitters vs placebo.

- T-cell levels over time were examined in anti-TNF–naive and anti-TNF–experienced patients who received etrolizumab or placebo **(Figures 7 and 8)**
 - There was a trend toward increased levels of all T-cell subsets during etrolizumab treatment in anti-TNF–naive remitters compared with either anti-TNF–naive non-remitters or placebo-treated patients **(Figure 7A-C)**
 - Peripheral blood CD3+ T-cell levels in patients with UC were lower than in healthy controls at baseline **(Figure 7D)**
 - Anti-TNF–naive patients who achieved remission with etrolizumab demonstrated an increase in peripheral blood CD3+ T cells at day 71 to levels comparable with those observed in healthy controls
 - In contrast, CD3+ T-cell levels in placebo-treated patients and etrolizumab non-remitters did not change during treatment

Figure 7. Levels of (A) CD3+ T Cells, (B) Treg and (C) Th17 Cells Were Increased in Anti-TNF–Naive Etrolizumab Remitters, and (D) CD3+ T Cells in Anti-TNF–Naive Etrolizumab Remitters Returned to Levels Comparable with Healthy Controls at Day 71

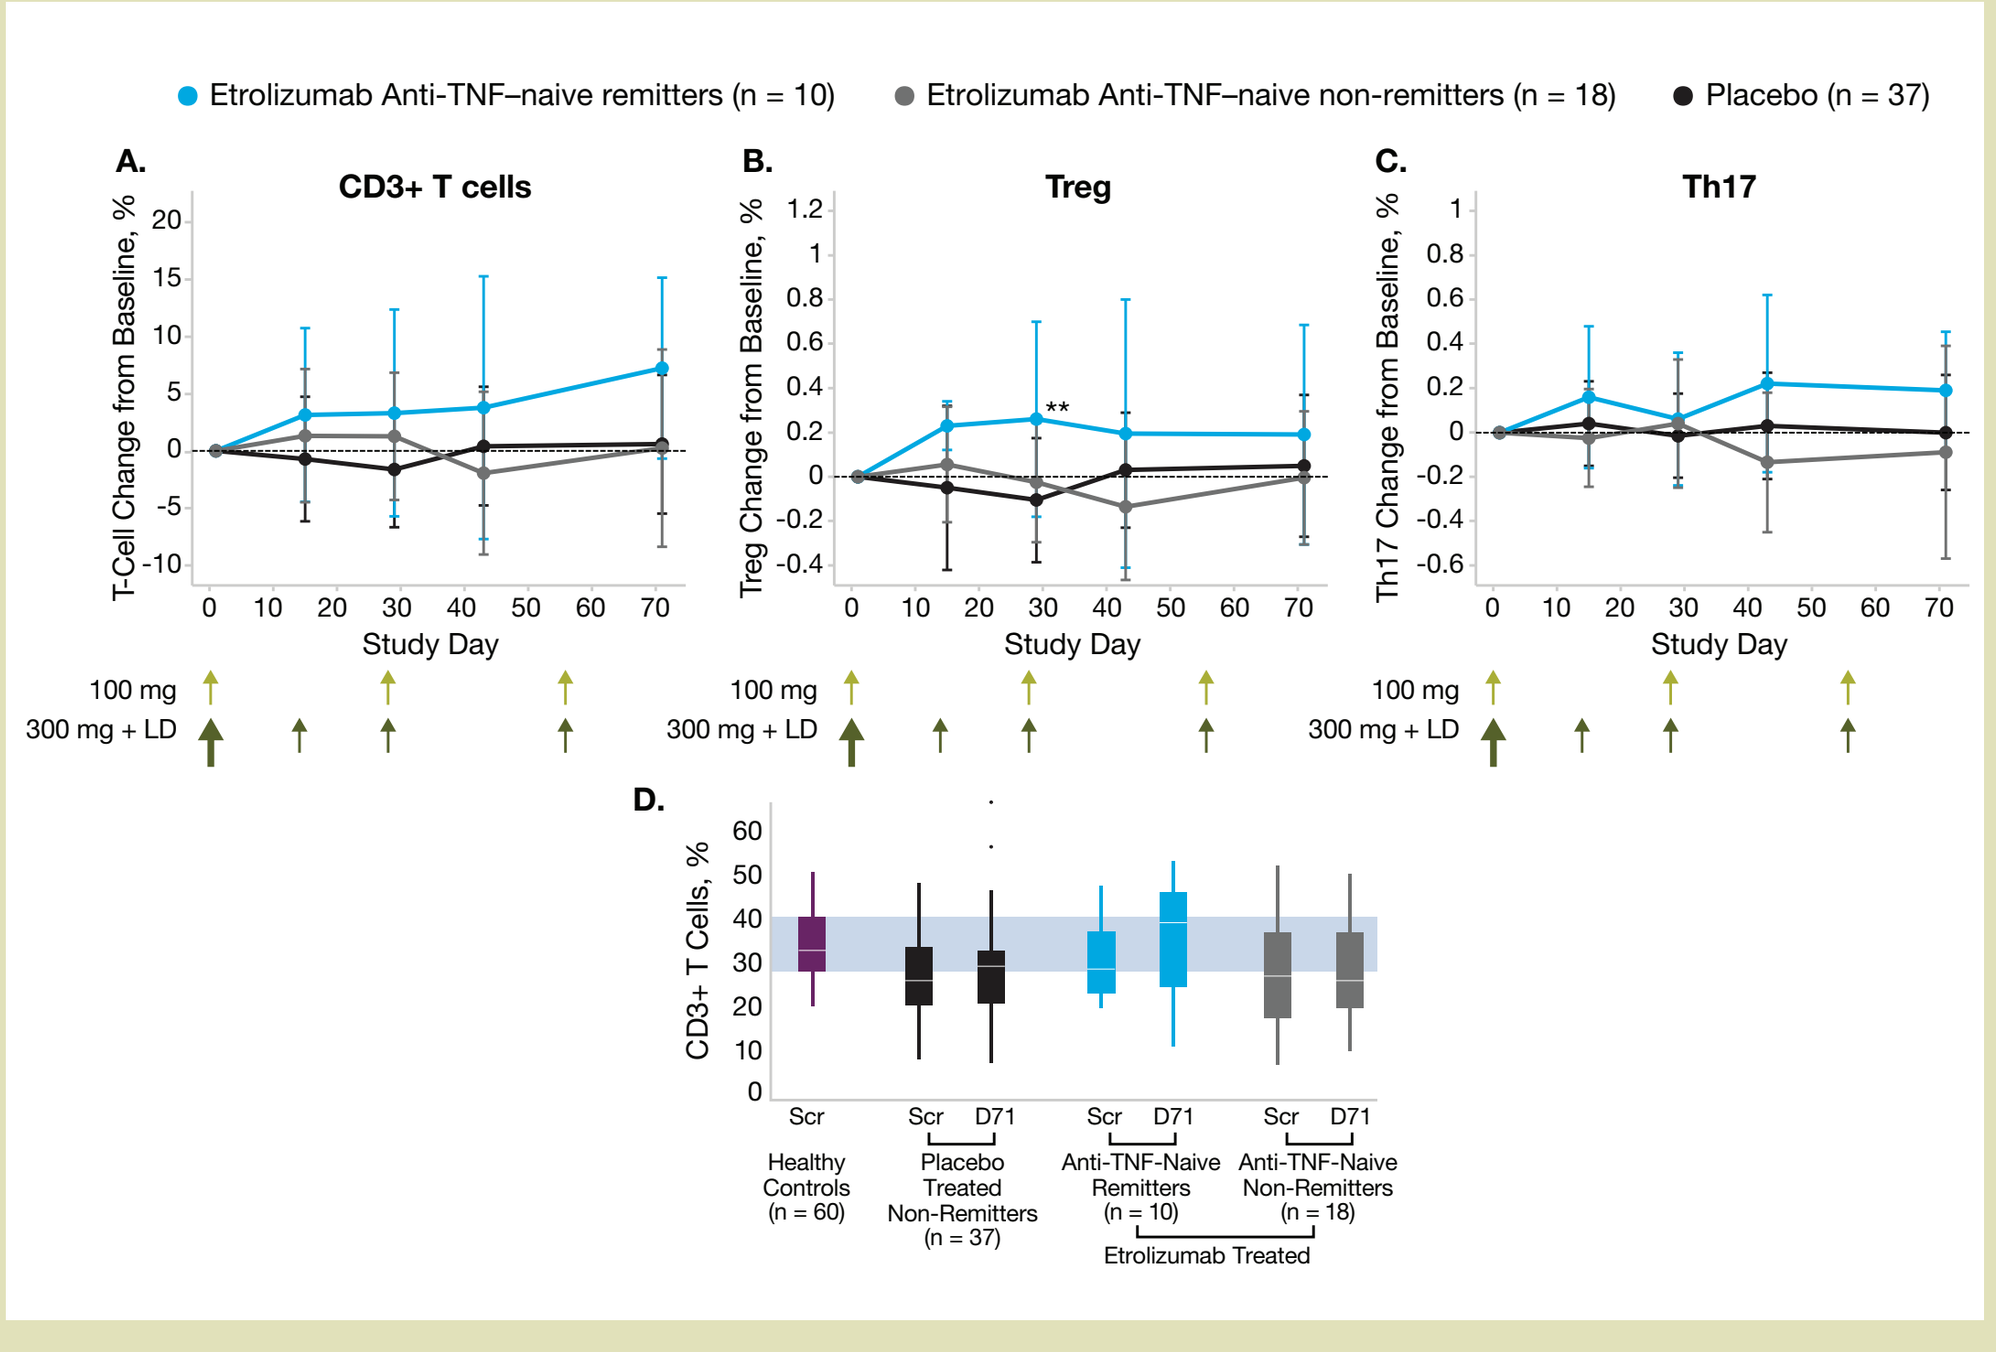

Etrolizumab Anti-TNF–naive remitters (n = 10)
Etrolizumab Anti-TNF–naive non-remitters (n = 18)
Placebo (n = 37)
A.
CD3+ T cells
T-Cell Change from Baseline, %
B.
Treg
Treg Change from Baseline, %
C.
Th17
Th17 Change from Baseline, %
Study Day
100 mg
300 mg + LD
D.
CD3+ T Cells, %
Scr
D71
Healthy Controls (n = 60)
Placebo Treated Non-Remitters (n = 37)
Anti-TNF-Naive Remitters (n = 10)
Anti-TNF-Naive Non-Remitters (n = 18)
Etrolizumab Treated

LD, loading dose; Scr, screening; Th17, T helper 17 cell; TNF, tumor necrosis factor; Treg, regulatory T cell. Data are expressed as group median ± median absolute deviation (A, B, C). There were no remitters in the placebo arm. **$P \leq 0.02$ for etrolizumab-treated remitters vs placebo. Data are expressed as the percentage of CD3+ T cells of total peripheral blood leukocytes (D). Shaded area denotes range in healthy controls.

- A similar trend toward increased levels of T-cell subsets was also observed in anti-TNF–experienced patients, although the number of etrolizumab remitters in this group was small (n = 3) **(Figure 8A-C)**
 - Levels of CD3+ T cells in anti-TNF–experienced etrolizumab remitters also increased to normal levels from screening to day 71 **(Figure 8D)**
- The median increase in CD3+ T cells appeared more profound in etrolizumab remitters who were anti-TNF experienced than in those who were anti-TNF naive (2.9-fold and 1.27-fold, respectively, at day 71)

Figure 8. Levels of (A) CD3+ T Cells, (B) Treg and (C) Th17 Cells Were Increased in Anti-TNF–Experienced Etrolizumab Remitters, and (D) CD3+ T Cells in Anti-TNF–Experienced Etrolizumab Remitters Returned to Levels Comparable with Healthy Controls at Day 71

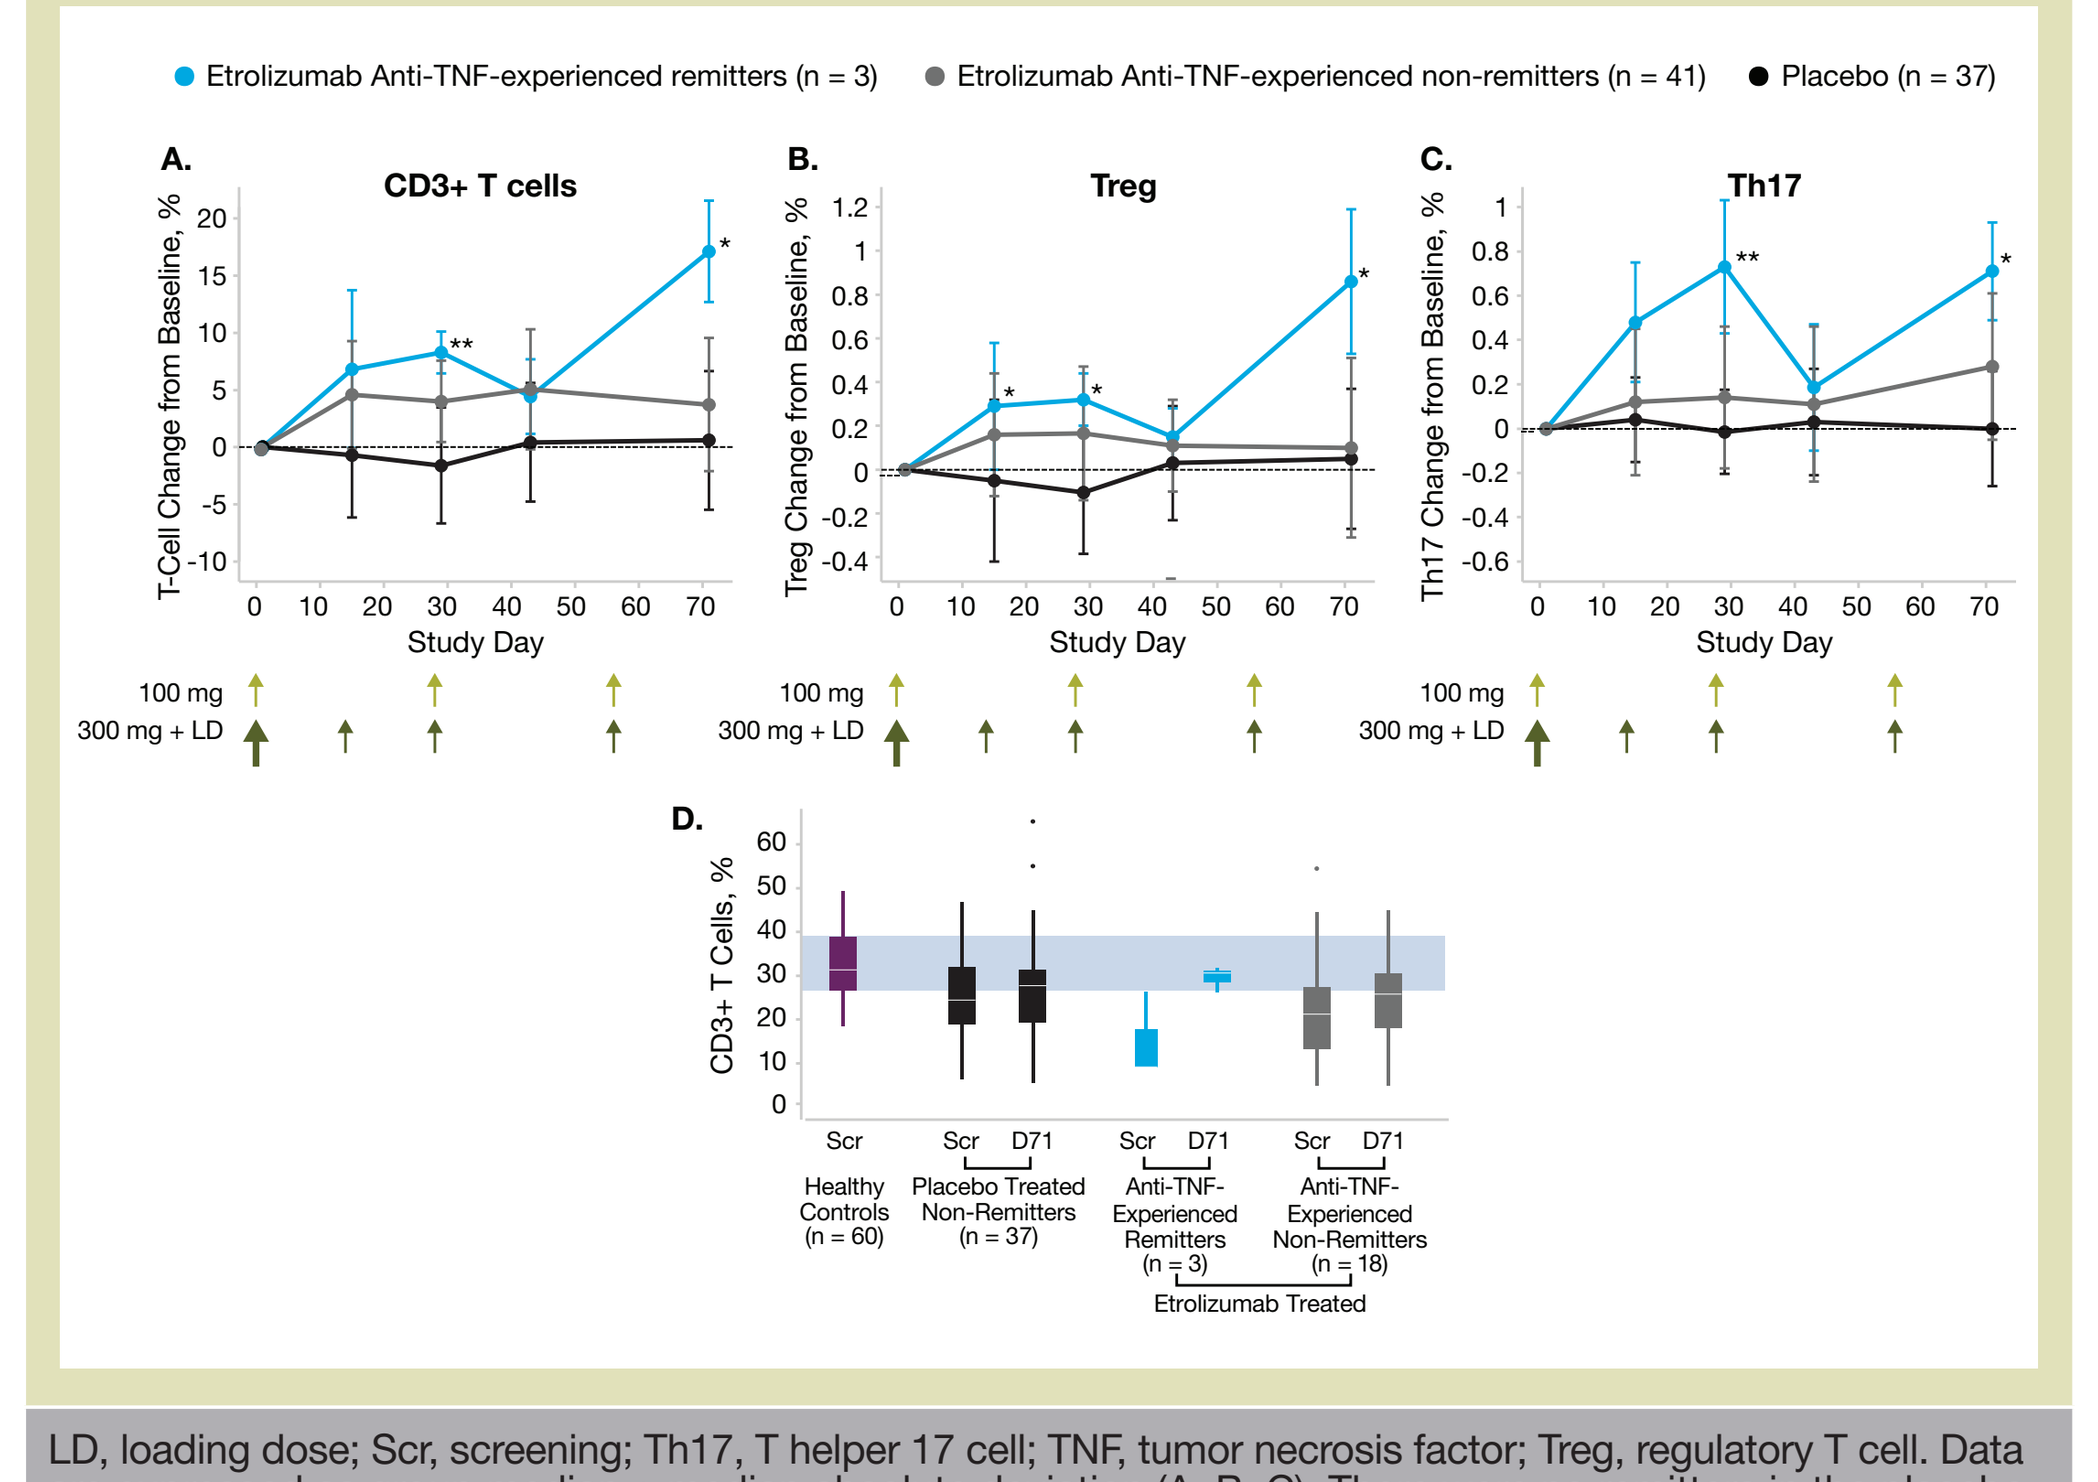

Etrolizumab Anti-TNF-experienced remitters (n = 3)
Etrolizumab Anti-TNF-experienced non-remitters (n = 41)
Placebo (n = 37)
A.
CD3+ T cells
T-Cell Change from Baseline, %
B.
Treg
Treg Change from Baseline, %
C.
Th17
Th17 Change from Baseline, %
Study Day
100 mg
300 mg + LD
D.
CD3+ T Cells, %
Scr
D71
Healthy Controls (n = 60)
Placebo Treated Non-Remitters (n = 37)
Anti-TNF-Experienced Remitters (n = 3)
Anti-TNF-Experienced Non-Remitters (n = 18)
Etrolizumab Treated

LD, loading dose; Scr, screening; Th17, T helper 17 cell; TNF, tumor necrosis factor; Treg, regulatory T cell. Data are expressed as group median ± median absolute deviation (A, B, C). There were no remitters in the placebo arm. *$P \leq 0.05$ and **$P \leq 0.02$ for etrolizumab-treated remitters vs placebo. Data are expressed as the percentage of CD3+ T cells of total peripheral blood leukocytes (D). Shaded area denotes range in healthy controls.

- Treg levels at baseline were higher in anti-TNF–naive etrolizumab remitters than in anti-TNF–naive non-remitters or placebo-treated patients, and in fact were within the range of Treg levels in healthy controls **(Figure 9A)**
- Stratification of Treg levels at baseline suggests that a higher percentage of Treg at baseline may be associated with patient remission upon etrolizumab treatment **(Figure 9B)**

Figure 9. Potential Predictive Value of Patient Remission by Treg Levels at Baseline

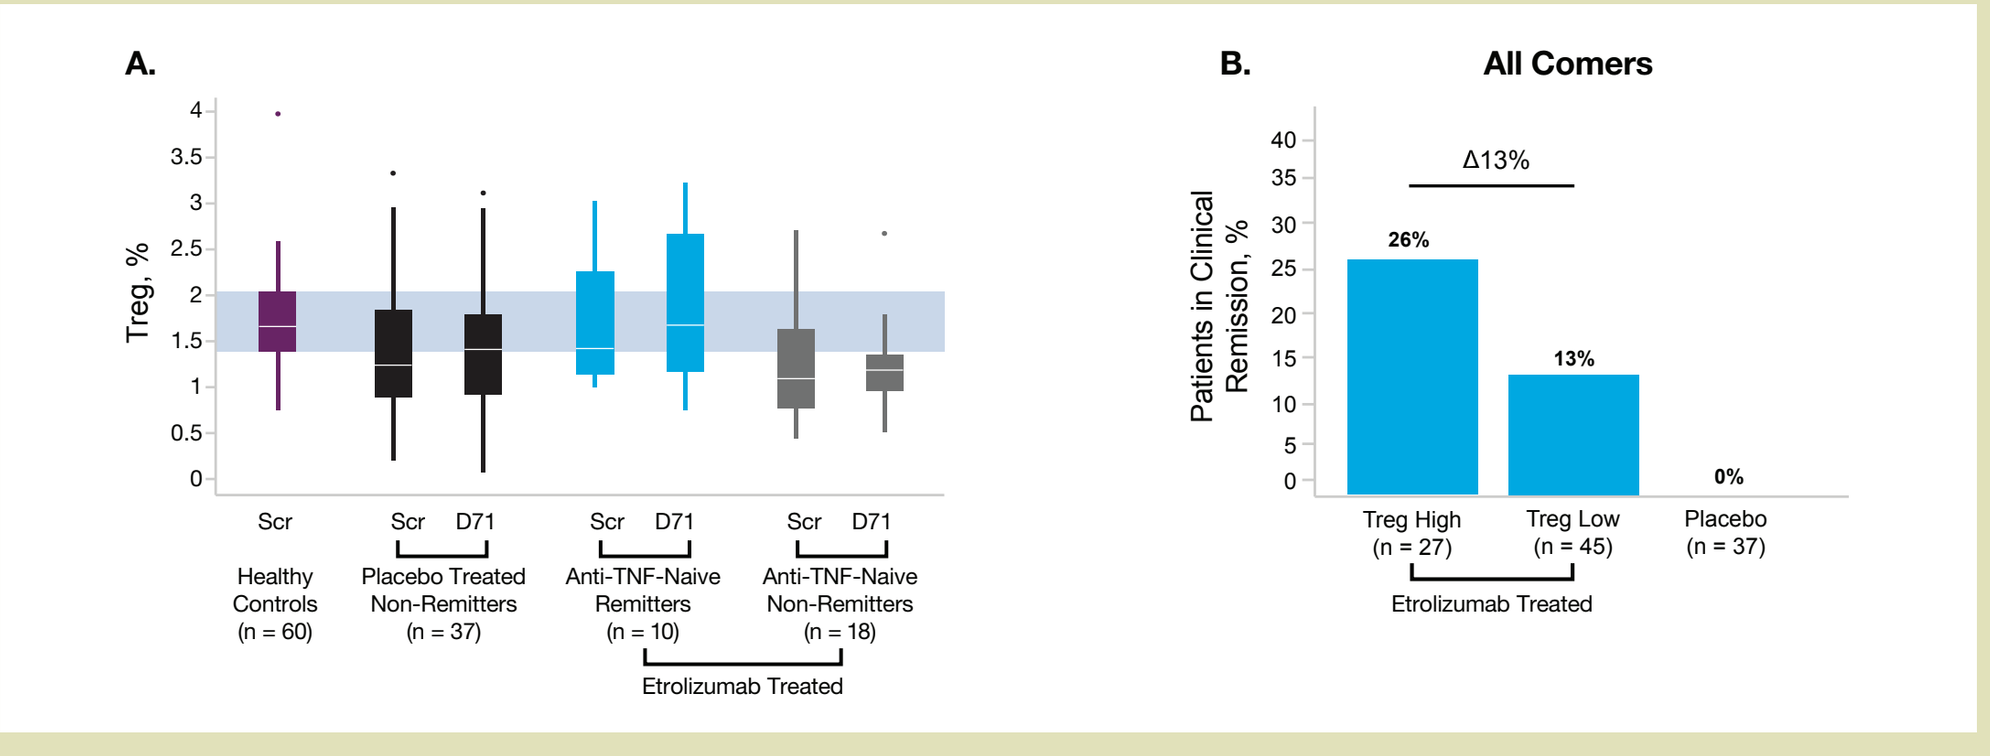

A.
Treg, %
Scr
D71
Healthy Controls (n = 60)
Placebo Treated Non-Remitters (n = 37)
Anti-TNF-Naive Remitters (n = 10)
Anti-TNF-Naive Non-Remitters (n = 18)
Etrolizumab Treated
B.
All Comers
Patients in Clinical Remission, %
Δ13%
26%
13%
0%
Treg High (n = 27)
Treg Low (n = 45)
Placebo (n = 37)
Etrolizumab Treated

Scr, screening; TNF, tumor necrosis factor; Treg, regulatory T cell. There were no remitters in the placebo arm. Shaded area denotes range in healthy controls. Data are expressed as the percentage of Treg of total peripheral blood leukocytes (A). Median of baseline Treg percentage of 1.28 was used as cutoff. A value of ≥ 1.28 was considered "Treg High" and < 1.28 was considered "Treg Low" (B).

CONCLUSIONS

- Using a novel epigenetic approach, we demonstrated that patients with UC had lower baseline peripheral blood CD3+ T-cell and Treg levels compared with healthy donors, especially anti-TNF–experienced patients with UC
 - These data indicate that patients with UC have an altered composition of peripheral blood T cells
- Etrolizumab treatment was associated with increased levels of total T cells, Treg and Th17 cells in peripheral blood
 - Among etrolizumab-treated patients, more pronounced increases in T-cell subsets were observed in patients who achieved clinical remission than in those who did not
- Reduced levels of peripheral CD3+ T cells at baseline observed in patients with UC appeared to normalize in etrolizumab-treated patients who achieved remission
- Higher levels of Treg at baseline may be associated with clinical remission upon etrolizumab treatment
 - These data suggest that peripheral blood Treg could be a biomarker to identify patients who may experience enhanced clinical benefit from etrolizumab

REFERENCES

1. Marsal J, et al. *J Intern Med.* 2012;272(5):411-429.
2. Vermeire S, et al. *Lancet.* 2014;384(9940):309-318.
3. Geremia A, et al. *Autoimmunity Rev.* 2014;13(1):3-10.
4. Wieczorek G, et al. *Cancer Res.* 2009;69(2):599-608.
5. Sehouli J, et al. *Epigenetics.* 2011;6(2):236-246.
6. Singh A, et al. *Allergy Asthma Clin Immunol.* 2014;10(1):32.
7. Baron U, et al. *Eur J Immunol.* 2007;37(9):2378-2389.

DISCLOSURES

Franklin Fuh, Akiko Chai, Caroline Looney, Sharon O'Byrne and **Teresa Ramirez-Montagut** are employees of Genentech, Inc., a member of the Roche group. **Eva Raschke** and **Thomas O. Kleen** are employees of Epiontis GmbH. **Séverine Vermeire** has received compensation for consulting from AstraZeneca and Ferring; received grant/research support from MSD, Takeda and AbbVie; has served on advisory committees or review panels for Ferring, MSD, Pfizer, Shire, AbbVie, Mundipharma, Hospira, Takeda and Genentech, Inc., a member of the Roche group; and received speaker fees from Ferring, MSD, Pfizer, AbbVie, Hospira and Takeda.

https://www.etroposters.com/index.cfm?iEventID=1&iFileID=147

Presented at United European Gastroenterology Week; Oct 24-28, 2015; Barcelona, Spain

This study was funded by Genentech, Inc., a member of the Roche Group.
Support for third-party writing assistance for this poster was provided by F. Hoffmann-La Roche Ltd.