Validation of BTK & Aiolos Immunophenotyping Assay to Monitor B, NK & T cell Responses as a Clinical Biomarker in B-cell Malignancies

CYTO 2021

Abstract#254

Jackie Benko[1], May Tan[2], Angelina Bisconte [1], Natalya Belkina PhD [1], Deborah Phippard PhD[1], Janine Powers PhD[2]

[1]Precision for Medicine, Frederick, Maryland; [2]Nurix Therapeutics, Inc., San Francisco, California

INTRODUCTION

Precision for Medicine developed and validated a flow immunophenotyping assay to assess modulation of Bruton's tyrosine kinase (BTK) and Aiolos in whole blood, which will be used as a pharmacodynamic biomarker for Nurix Therapeutic's clinical studies. Nurix's small molecule NX-2127, a BTK chimeric targeting molecule, is a dual protein degrader of BTK and Aiolos. This therapeutic candidate is intended for treatment of patients with B-cell malignancies. Here, we validated a fit for purpose 10-color flow cytometry assay to quantify the degradation of BTK and Aiolos protein in B, NK, and T cells in peripheral whole blood using Streck Cyto-Chex® BCT tubes.

PURPOSE

Measuring modulation of BTK and Aiolos in B cells can be challenging in patients on standard of care therapies. This assay was validated to be both sensitive and selective in the quantification of BTK and Aiolos to facilitate the determination of an optimal therapeutic dose and correlate pharmacodynamic responses to potential clinical responses in the treatment of hematologic malignancies. The validation parameters assessed included intra-assay, inter-assay, inter-operator precision, and post-staining stability.

METHOD

Step 1. Whole Blood Collection in Cyto-Chex® BCT tubes
- Whole blood from 3 healthy donors were used to validate the assay.
- The blood was drawn directly into Cyto-Chex® BCT tubes and incubated at RT for 24hr prior to staining to ensure the cells were fixed by the proprietary fixative.

Step 2. Cell Surface Marker Staining
- Fixed whole blood was lysed and washed to remove red blood cells.
- Leukocytes were stained with an antibody cocktail containing cell surface markers for identification of T/B/NK cell subsets and Monocytes CD45, CD3, CD4, CD8, CD14, CD16, CD19 and CD56.

Step 3. Intracellular Staining
- Cells were permeabilized and stained with intracellular antibody cocktail containing BTK and Aiolos, then washed and stained with fluorescently labeled secondary IgG antibody to detect BTK.
- Sample was resuspended in staining buffer and transferred to BD TruCount™ tubes to acquire absolute counts of cell subsets of interest and for determining sensitivity.

Step 4. Cells were acquired on a BD LSRFortessa™ Cytometer

Step 5. Data analysis of BTK and Aiolos
- Data was analyzed with TreeStar's FlowJo™ software.
- % Frequency of Parent (FoP) T, B, NK and Monocyte cell populations were reported.
- BTK and Aiolos MFI and absolute cell counts in T cell, B cell, Monocyte, and NK cells were reported.

DETECTION OF BTK & AIOLOS

Figure 1. Detection of Intracellular BTK and Aiolos:

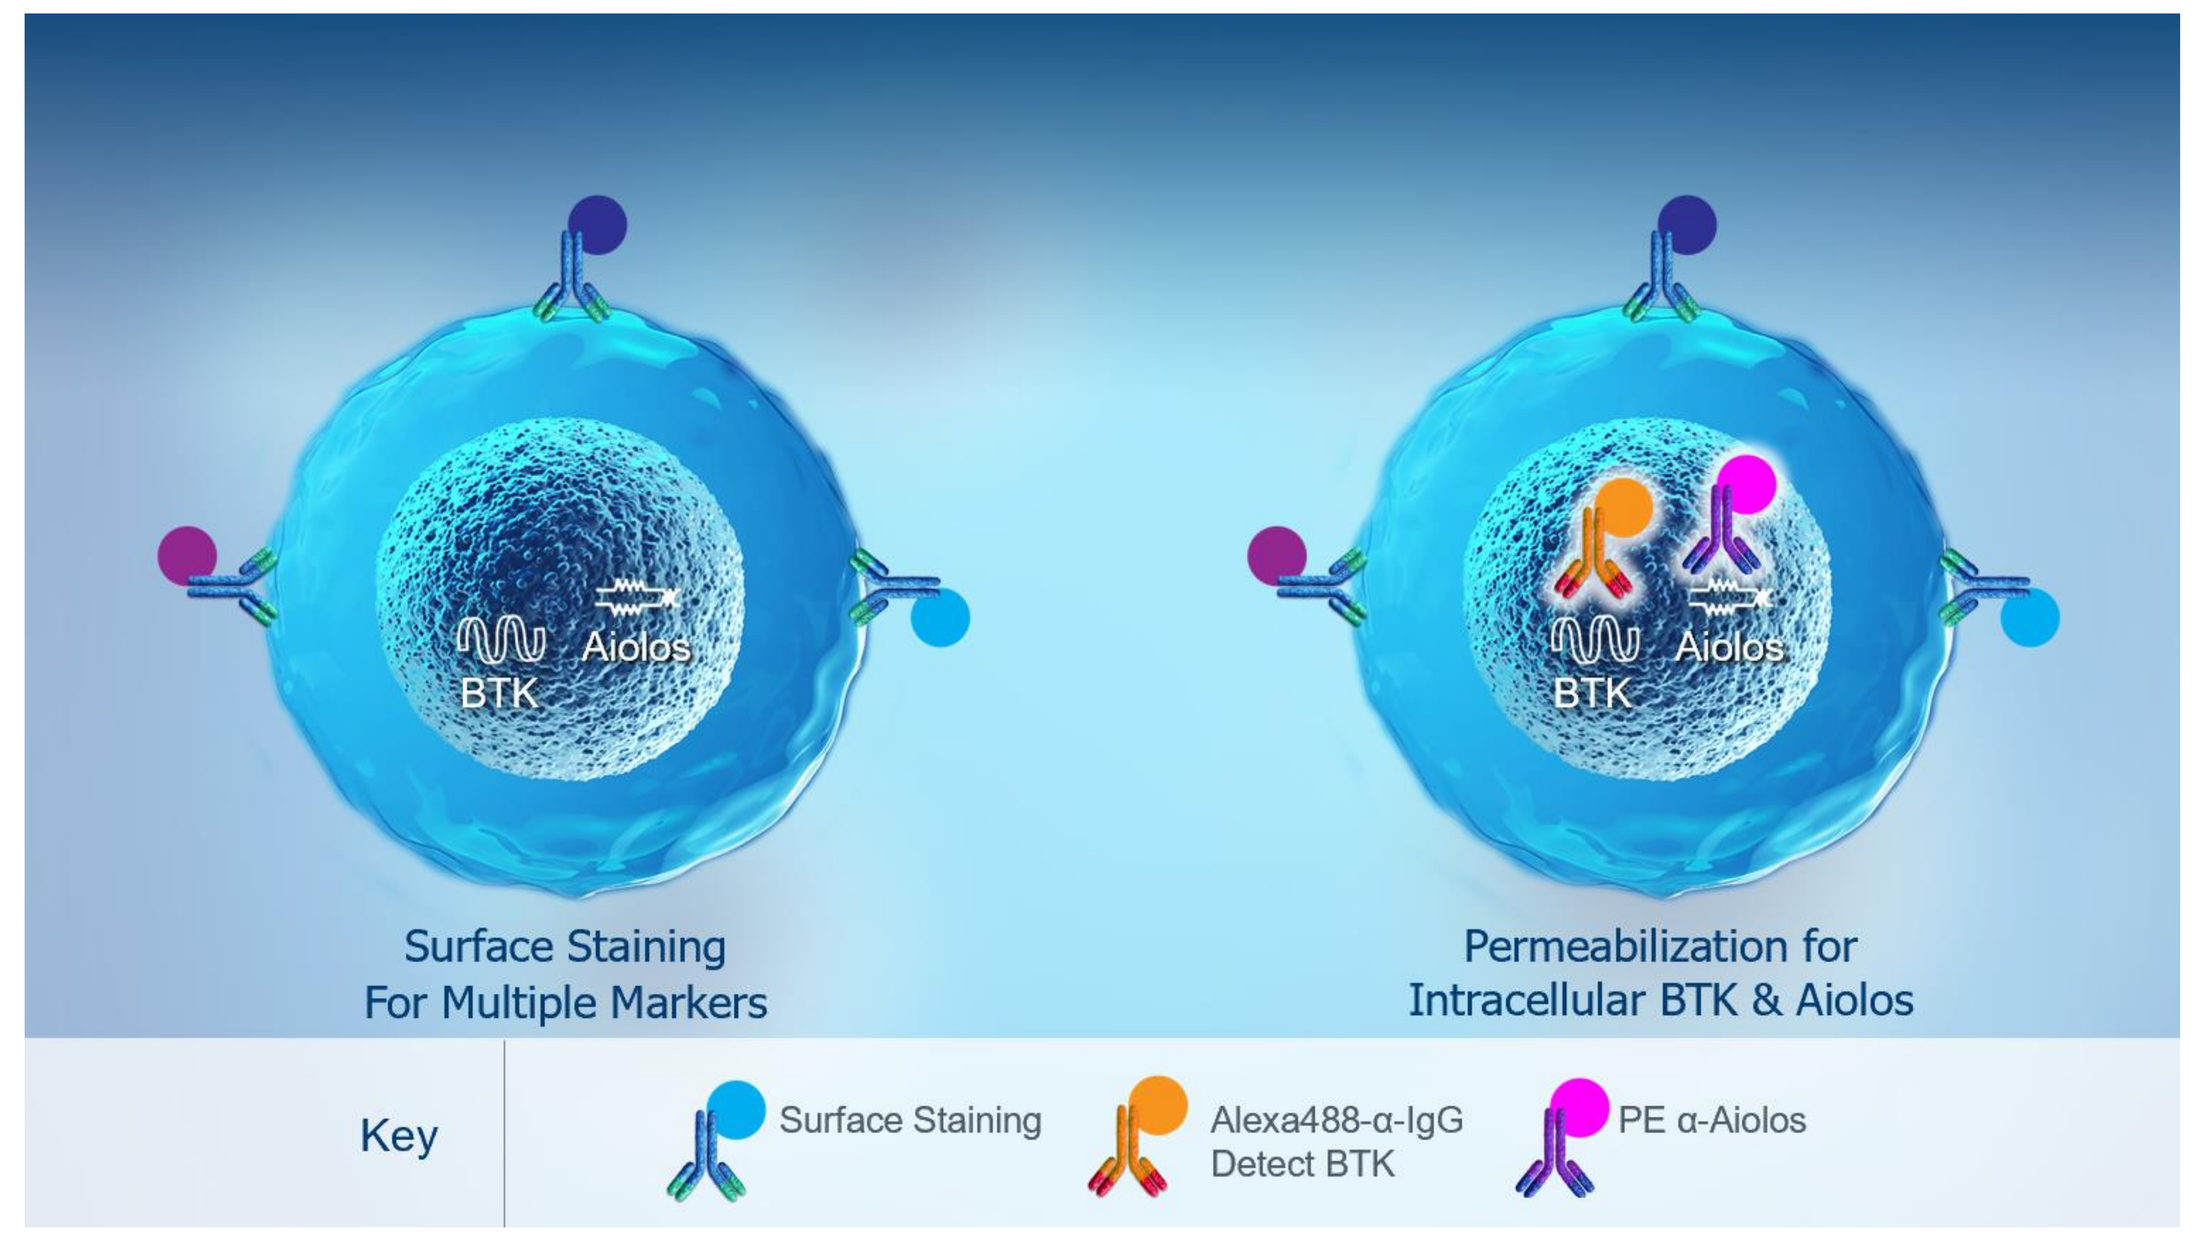
BTK
Aiolos
Surface Staining
For Multiple Markers
Permeabilization for
Intracellular BTK & Aiolos
Key
Surface Staining
Alexa488-α-IgG
Detect BTK
PE α-Aiolos

RESULTS

Summary of BTK and Aiolos Cyto-Chex® BCT Assay Validation
The assay was validated in whole blood with three healthy donors. Precision assessed the validation parameters of intra-assay, inter-assay, inter-operator and post-staining stability with pre-set assay validation criteria defined for each parameter being assessed.

Figure 2. Gating Strategy: CD4 & CD8 T cells, CD19+ B cells, and CD56+CD16+ NK cell subsets in whole blood

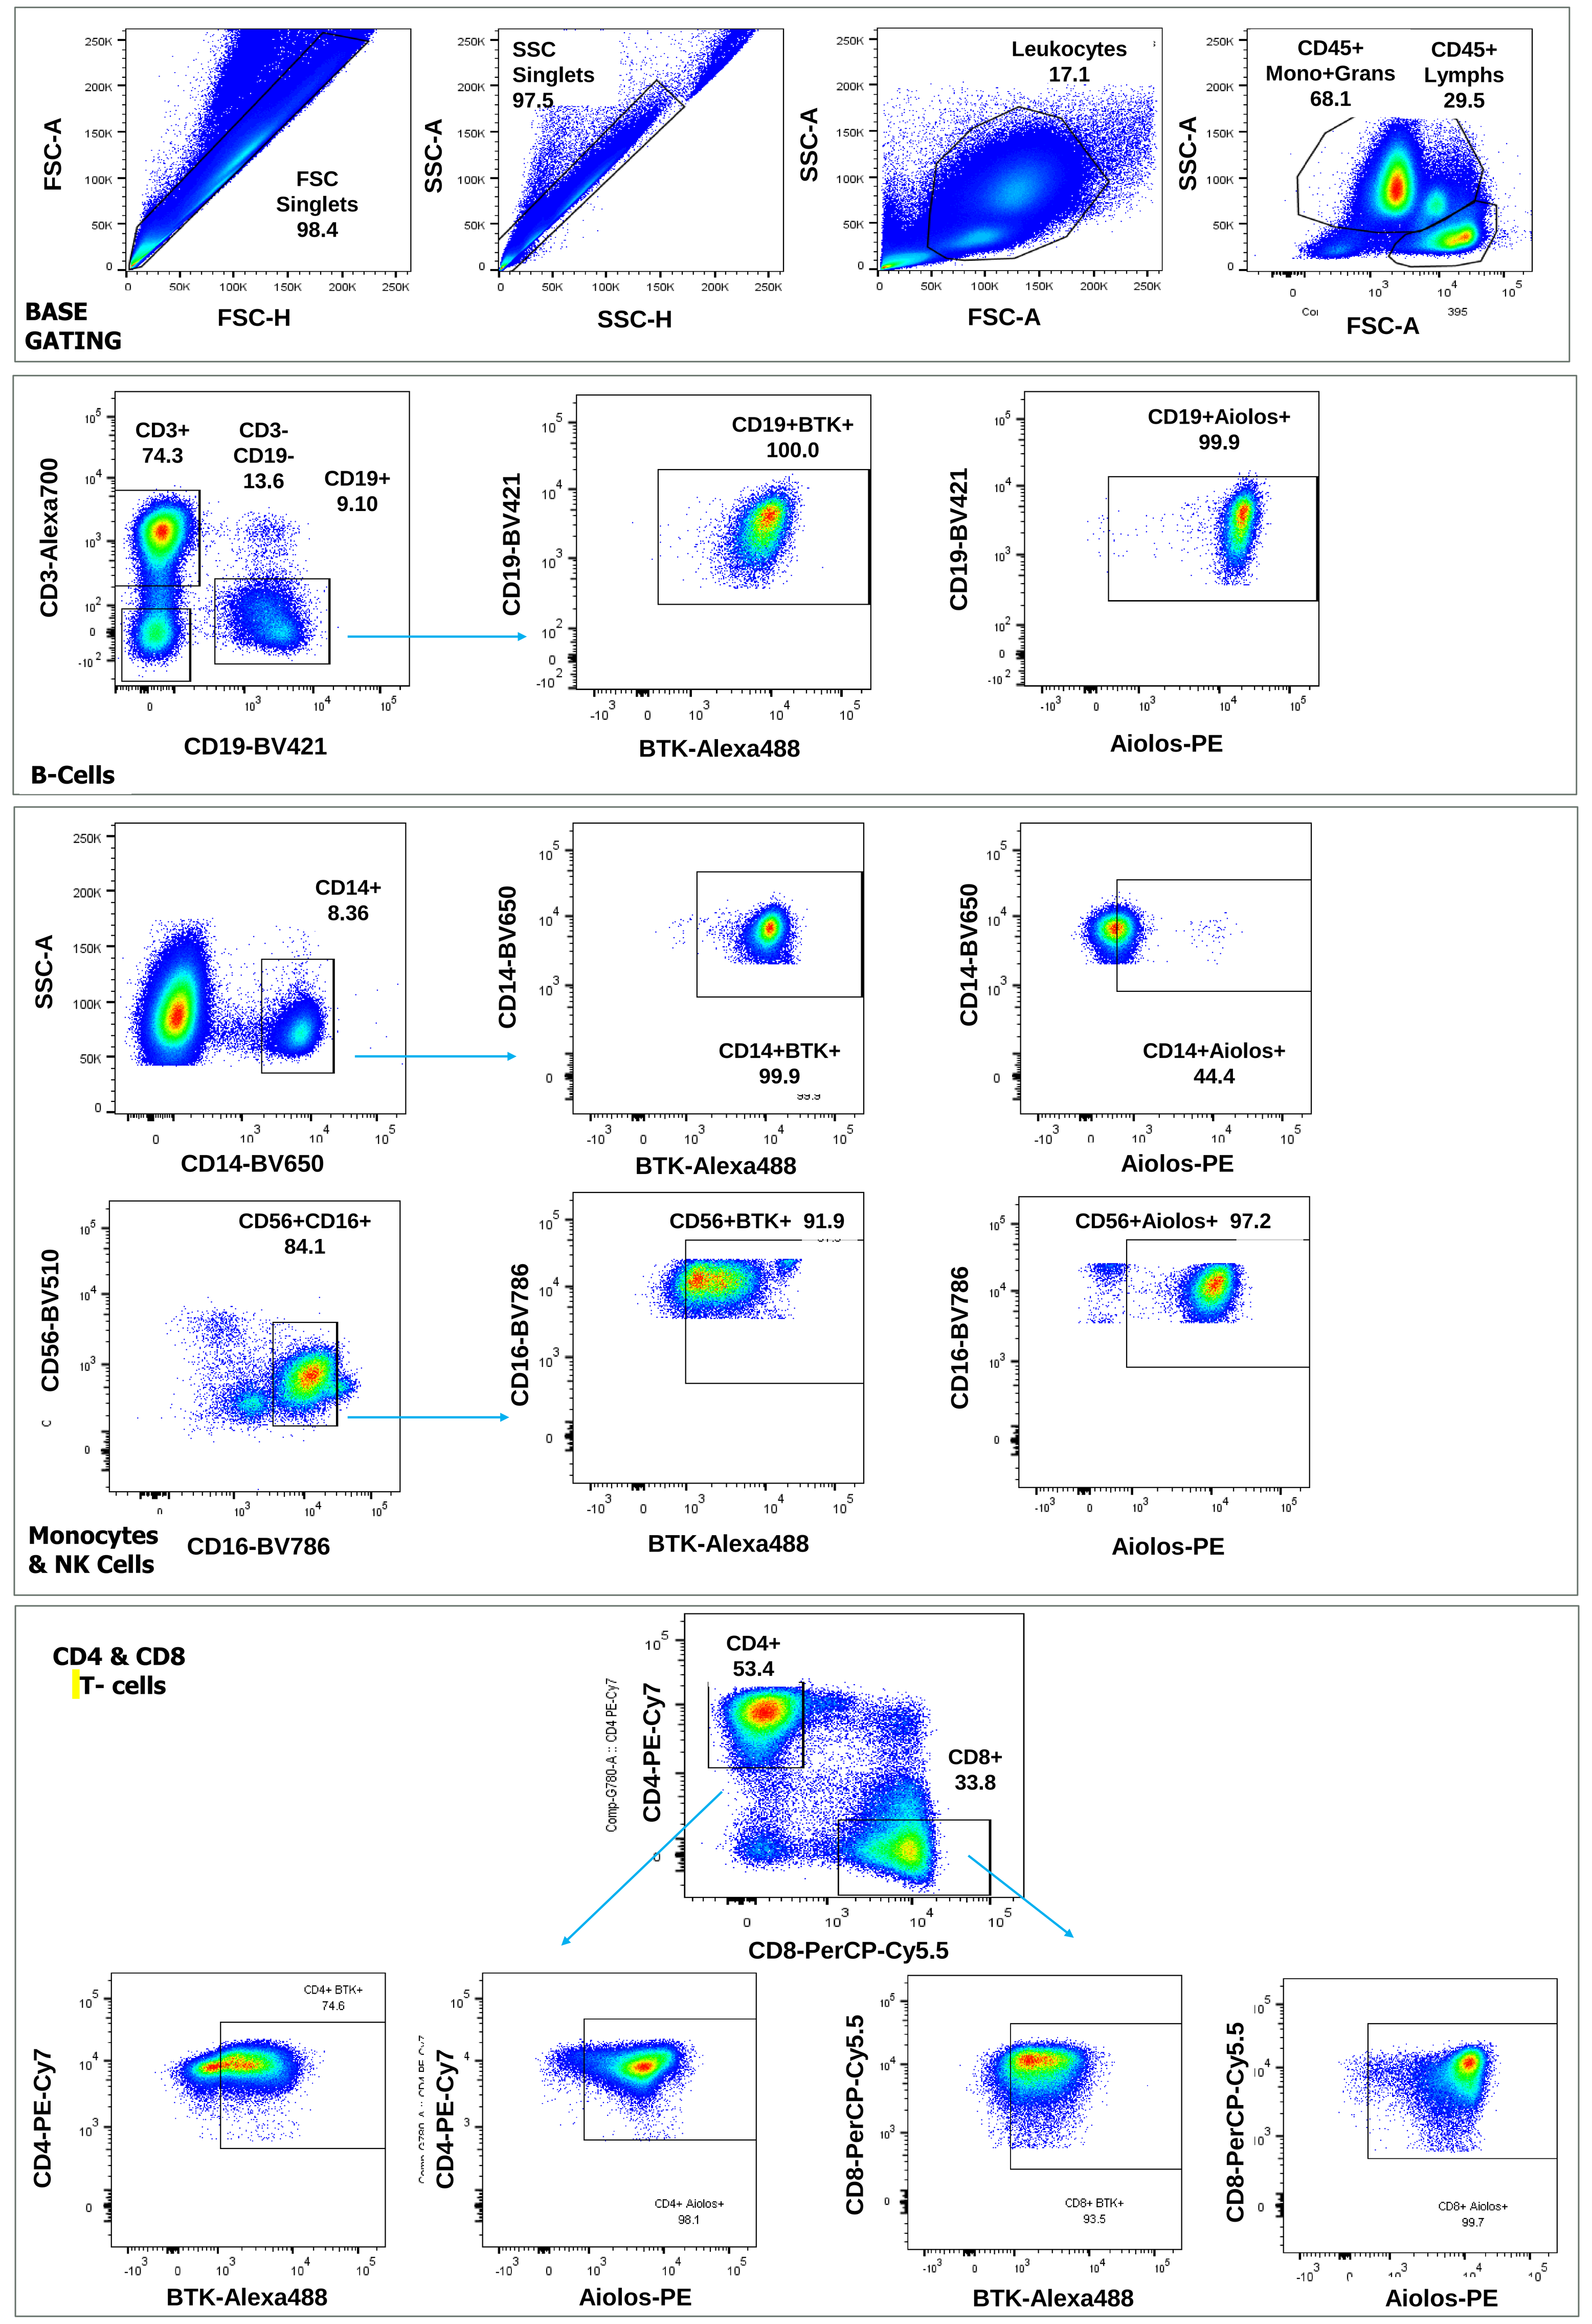
BASE
GATING
FSC Singlets
98.4
SSC Singlets
97.5
Leukocytes
17.1
CD45+
Mono+Grans
68.1
CD45+
Lymphs
29.5
FSC-A
FSC-H
SSC-A
SSC-H
FSC-A
B-Cells
CD3+
74.3
CD3-
CD19-
13.6
CD19+
9.10
CD3-Alexa700
CD19-BV421
CD19+BTK+
100.0
CD19+Aiolos+
99.9
BTK-Alexa488
Aiolos-PE
CD14+
8.36
CD14-BV650
CD14+BTK+
99.9
CD14+Aiolos+
44.4
Monocytes
& NK Cells
CD56+CD16+
84.1
CD56-BV510
CD16-BV786
CD56+BTK+ 91.9
CD56+Aiolos+ 97.2
CD4 & CD8
T- cells
CD4+
53.4
CD8+
33.8
CD4-PE-Cy7
CD8-PerCP-Cy5.5

Figure 3. Geometric Mean Fluorescence BTK (MFI) : Data representative of the 3 healthy donors tested. Mean of triplicates plotted with %CV. CD4 and CD8 BTK MFIs were similar. (CD8 data not shown)

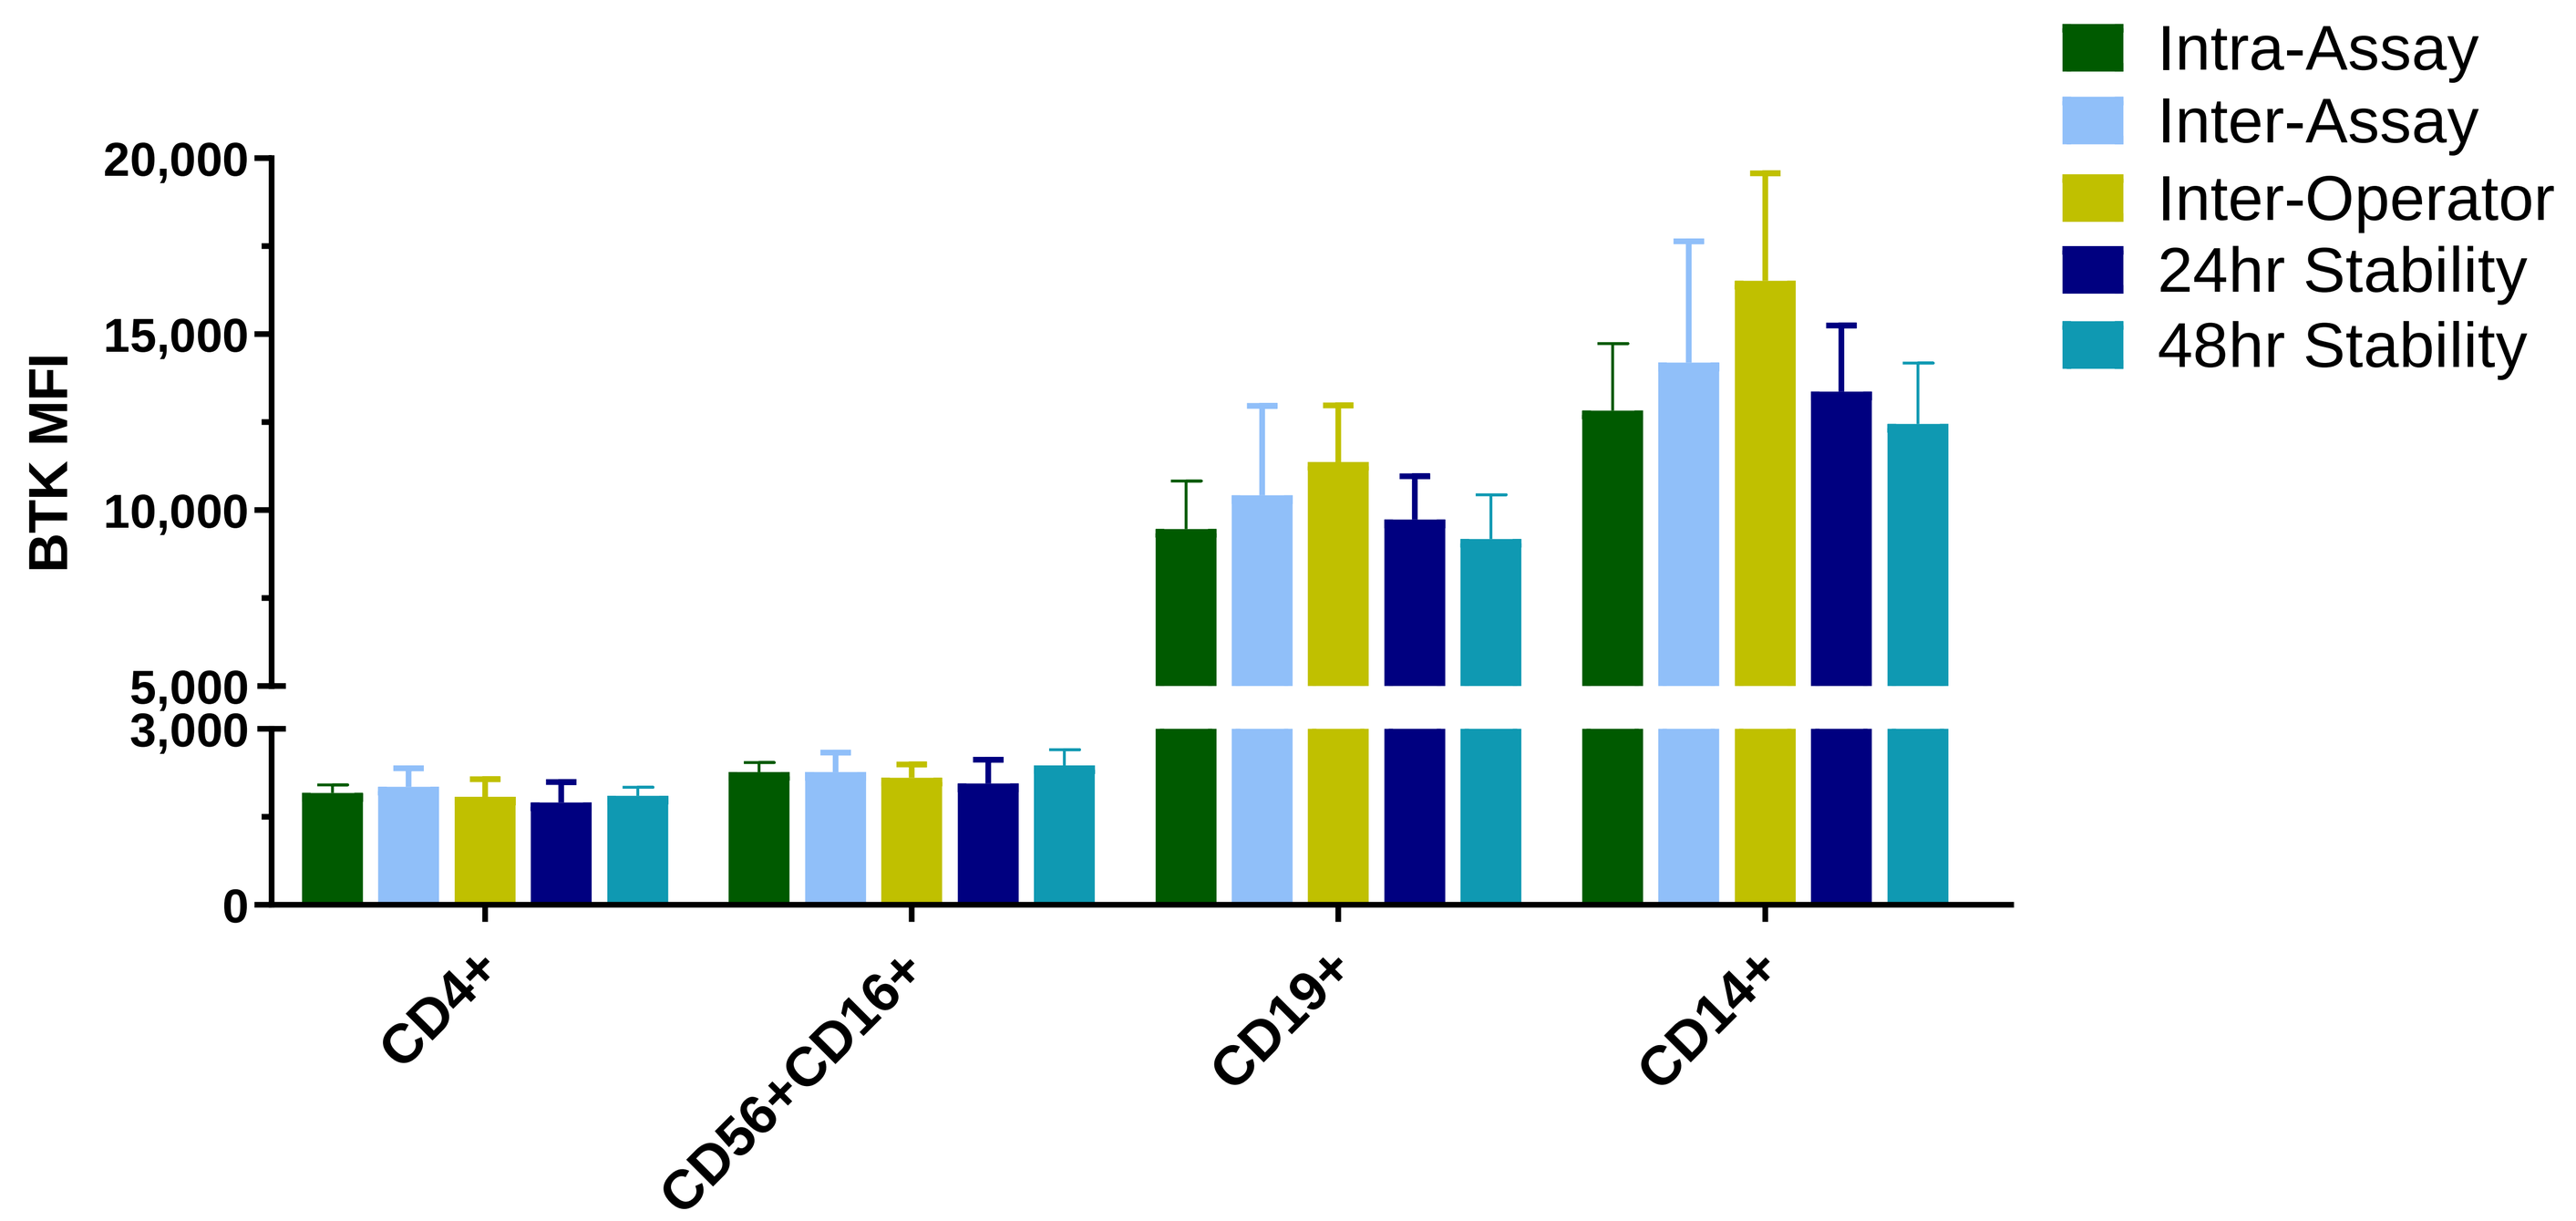
Intra-Assay
Inter-Assay
Inter-Operator
24hr Stability
48hr Stability
BTK MFI
CD4+
CD56+CD16+
CD19+
CD14+

Figure 4. Geometric Mean Fluorescence Aiolos+ (MFI) : Data representative of the 3 healthy donors tested. Mean of triplicates plotted with %CV. CD4 and CD8 BTK MFIs were similar. (CD8 data not shown)

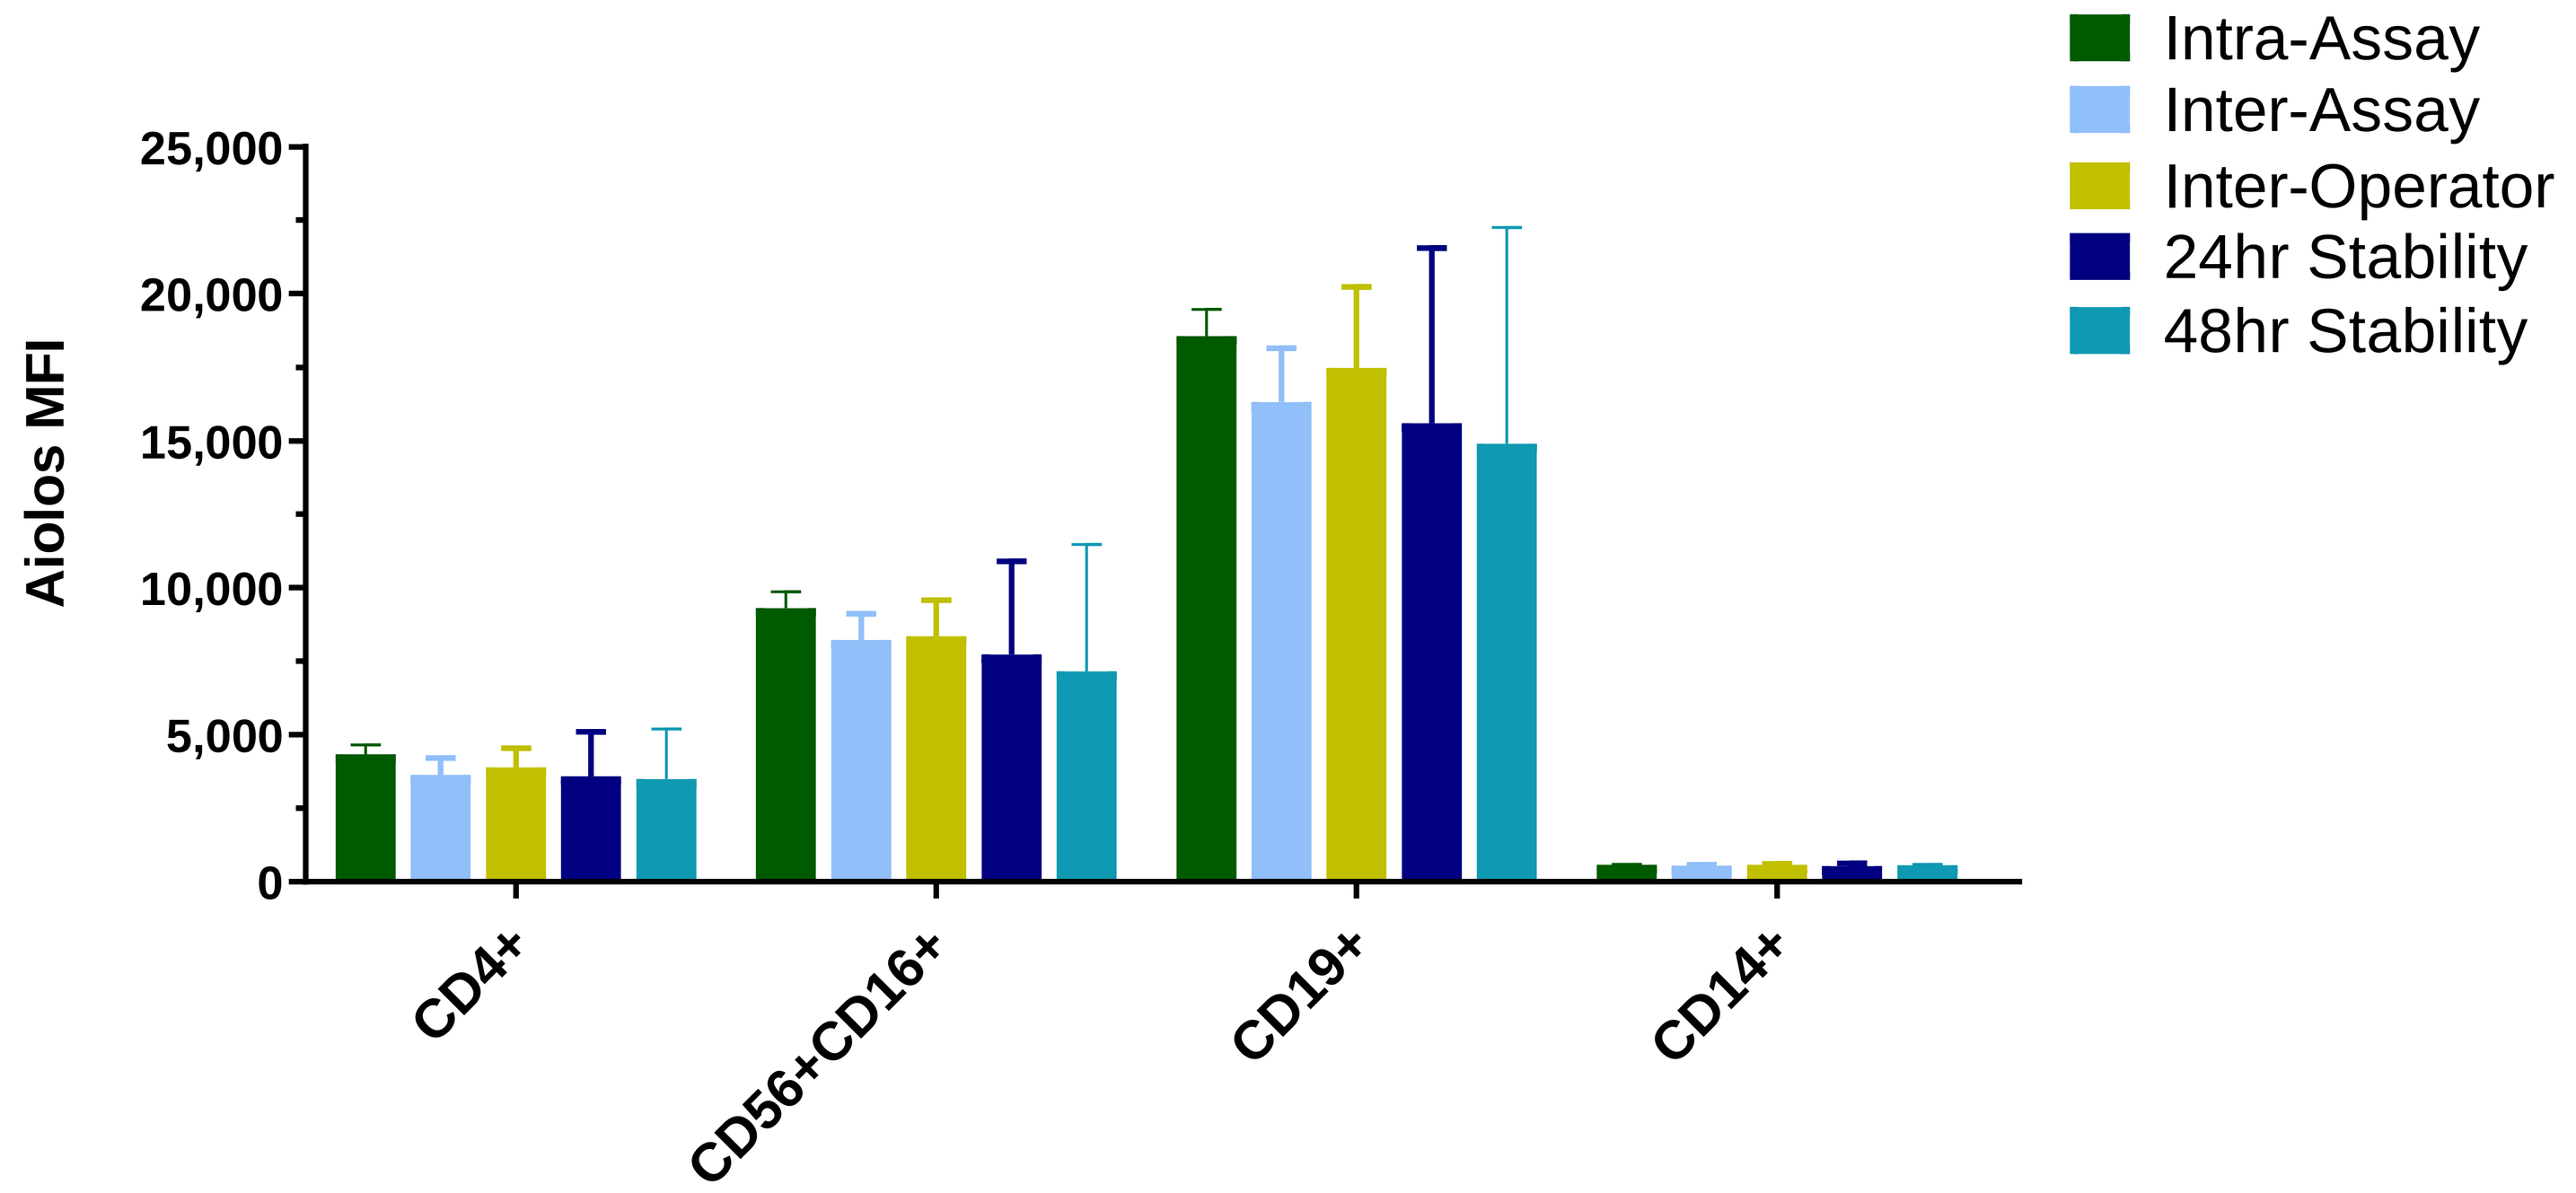
Intra-Assay
Inter-Assay
Inter-Operator
24hr Stability
48hr Stability
Aiolos MFI
CD4+
CD56+CD16+
CD19+
CD14+

Table 1. %Frequency of T/B/NK and Monocyte Cell Subsets Validation Results:
These results represent the Mean and %CV for the parameters tested in three healthy whole blood donors.

	Cell Population	Intra-Assay		Inter-Assay		Inter-Operator		24hr Post Staining		48hr Post Staining	
		Mean	%CV	Mean	%CV	Mean	%CV	Mean	%CV	Mean	%CV
Donor 1	CD4+	56.43	0.20	56.52	0.86	55.91	1.15	56.78	1.21	56.63	0.70
	CD8+	39.00	0.26	38.68	0.99	39.00	0.70	38.48	2.74	38.68	1.69
	CD56+ CD16+	81.37	0.43	81.07	0.82	76.9	10.69	79.45	4.84	83.75	5.70
	CD19+	8.67	0.64	8.38	5.49	8.45	7.27	8.82	3.51	8.19	11.59
	CD14+	5.62	0.71	5.60	0.94	5.56	7.92	5.66	1.53	5.63	0.73
Donor 2	CD4+	64.00	3.92	64.90	2.29	65.90	3.31	64.40	3.42	64.25	3.29
	CD8+	29.07	5.57	29.83	3.64	28.31	7.51	29.13	4.56	29.25	4.69
	CD56+ CD16+	90.30	3.08	87.20	4.85	80.23	21.58	88.78	4.28	89.55	3.04
	CD19+	8.28	4.68	8.54	9.20	8.27	12.91	8.69	10.04	8.63	8.78
	CD14+	5.74	4.56	5.52	9.21	5.39	10.51	5.76	3.76	5.72	3.84
Donor 3	CD4+	49.73	0.42	50.50	2.69	52.26	5.48	50.28	2.18	50.18	1.79
	CD8+	38.40	2.03	38.01	2.55	35.37	8.21	37.98	2.80	37.73	3.96
	CD56+ CD16+	78.93	15.05	77.87	14.94	55.61	39.19	79.55	12.29	76.15	14.69
	CD19+	11.83	1.76	11.98	3.80	12.26	7.31	12.00	3.12	11.90	1.82
	CD14+	7.47	3.71	7.41	2.69	7.37	2.72	7.43	3.24	7.38	4.07

RESULTS

Figure 5. Sensitivity of Absolute Cell Count of BTK in CD19+ B cells : Data representative of healthy donors tested, Mean and SD absolute counts of BTK in CD19+ cells and showed a and showed an R^2 linearity value of 0.9989. Eight dilutions of full stained cells (0, 1:2, 1:5, 1:10, 1:20, 1:40, 1:80, and 1:160) in 1 million PBMCs.

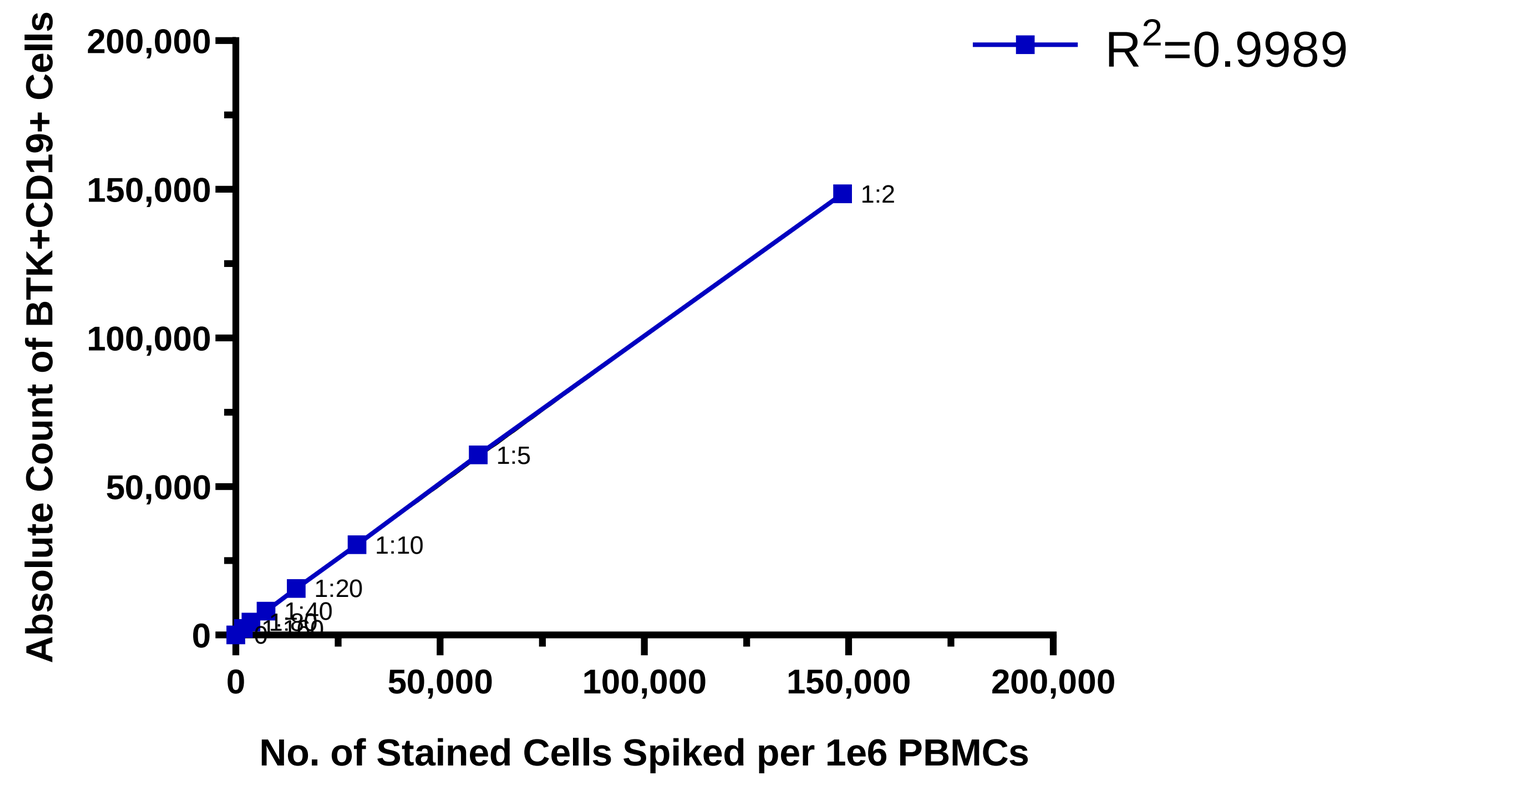
R2=0.9989
Absolute Count of BTK+CD19+ Cells
No. of Stained Cells Spiked per 1e6 PBMCs
1:2
1:5
1:10
1:20

Figure 6. Sensitivity of Absolute Cell Count of Aiolos in CD19+ B cells : Data representative of healthy donors tested, Mean and SD absolute counts of Aiolos in CD19+ cells and showed an R^2 linearity value of 0.9986. Eight dilutions of full stained cells (0, 1:2, 1:5, 1:10, 1:20, 1:40, 1:80, and 1:160) in 1 million PBMCs.

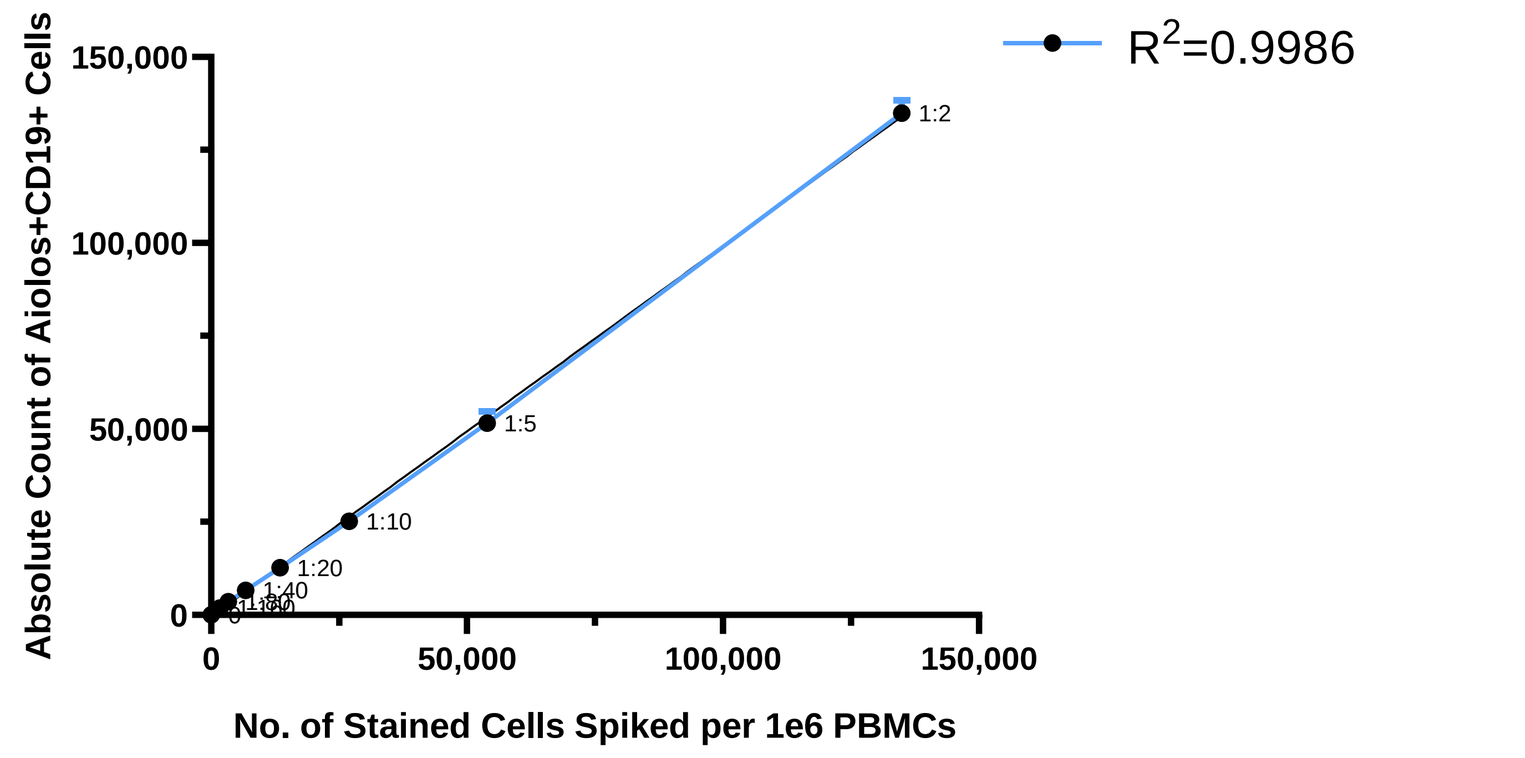
R2=0.9986
Absolute Count of Aiolos+CD19+ Cells
No. of Stained Cells Spiked per 1e6 PBMCs
1:2
1:5
1:10
1:20

CONCLUSION

This whole blood flow assay was successfully validated to quantify both BTK and Aiolos modulation in T, B, NK, and Monocyte cell subsets.

An observation of the validation is that the samples will need to be stained and acquired the same day because the post-staining stability results demonstrated a decrease in Aiolos MFI after 24hr if the samples are stained and stored at 4°C.

This assay can be used as a pharmacodynamic assay to facilitate the determination of an optimal therapeutic dose in treatment of B-cell malignancies.

ACKNOWLEDGEMENTS

We would like to extend our thanks and appreciation to the scientific, clinical and supportive teams at Nurix Therapeutics, Inc. and Precision for Medicine for their partnership and collaboration.

REFERENCES

Xia S et.al., Cellular & Molecular Immunology. 2020. 17:1042–1052.
Marcondes N et. al.,Clinical Cytometry Part B, 2018 94B:724.
Nisitani S et. al., Proc Natl Acad. Sci. 1997:2737–2742.

CONTACT INFORMATION

Jackie Benko
Email: Jackie.Benko@precisionformedicine.com